3932

A Phase 1 Dose-Escalation Study of Milademetan in Combination With 5-Azacitidine (AZA) in Patients With Acute Myeloid Leukemia (AML) or High-Risk Myelodysplastic Syndrome (MDS)

Courtney D. DiNardo,[1] Rebecca Olin,[2] Jo Ishizawa,[1] Hiroyuki Sumi,[3] Jingdong Xie,[4] Kazunobu Kato,[4] Prasanna Kumar,[4] Michael Andreeff[1]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Helen Diller Family Comprehensive Cancer Center, University of California, San Francisco, CA; [3]Daiichi Sankyo Co, Ltd, Tokyo, Japan; [4]Daiichi Sankyo, Inc, Basking Ridge, NJ

BACKGROUND

- Tumor suppressor protein p53, encoded by the *TP53* gene, is negatively regulated by murine double minute 2 (MDM2), an E3 ubiquitin ligase[1]
- Although *TP53* is infrequently mutated in AML, deregulation of MDM2 is commonly observed and results in aberrant degradation of p53, which prevents its tumor-suppressive functions[2]
- Milademetan (DS-3032b) is an oral, novel and specific small-molecule inhibitor of MDM2 that disrupts the interaction between MDM2 and p53 to prevent excessive degradation of p53 in MDM2-expressing tumors (**Figure 1**)[3]

Figure 1. Milademetan Proposed Mechanism of Action

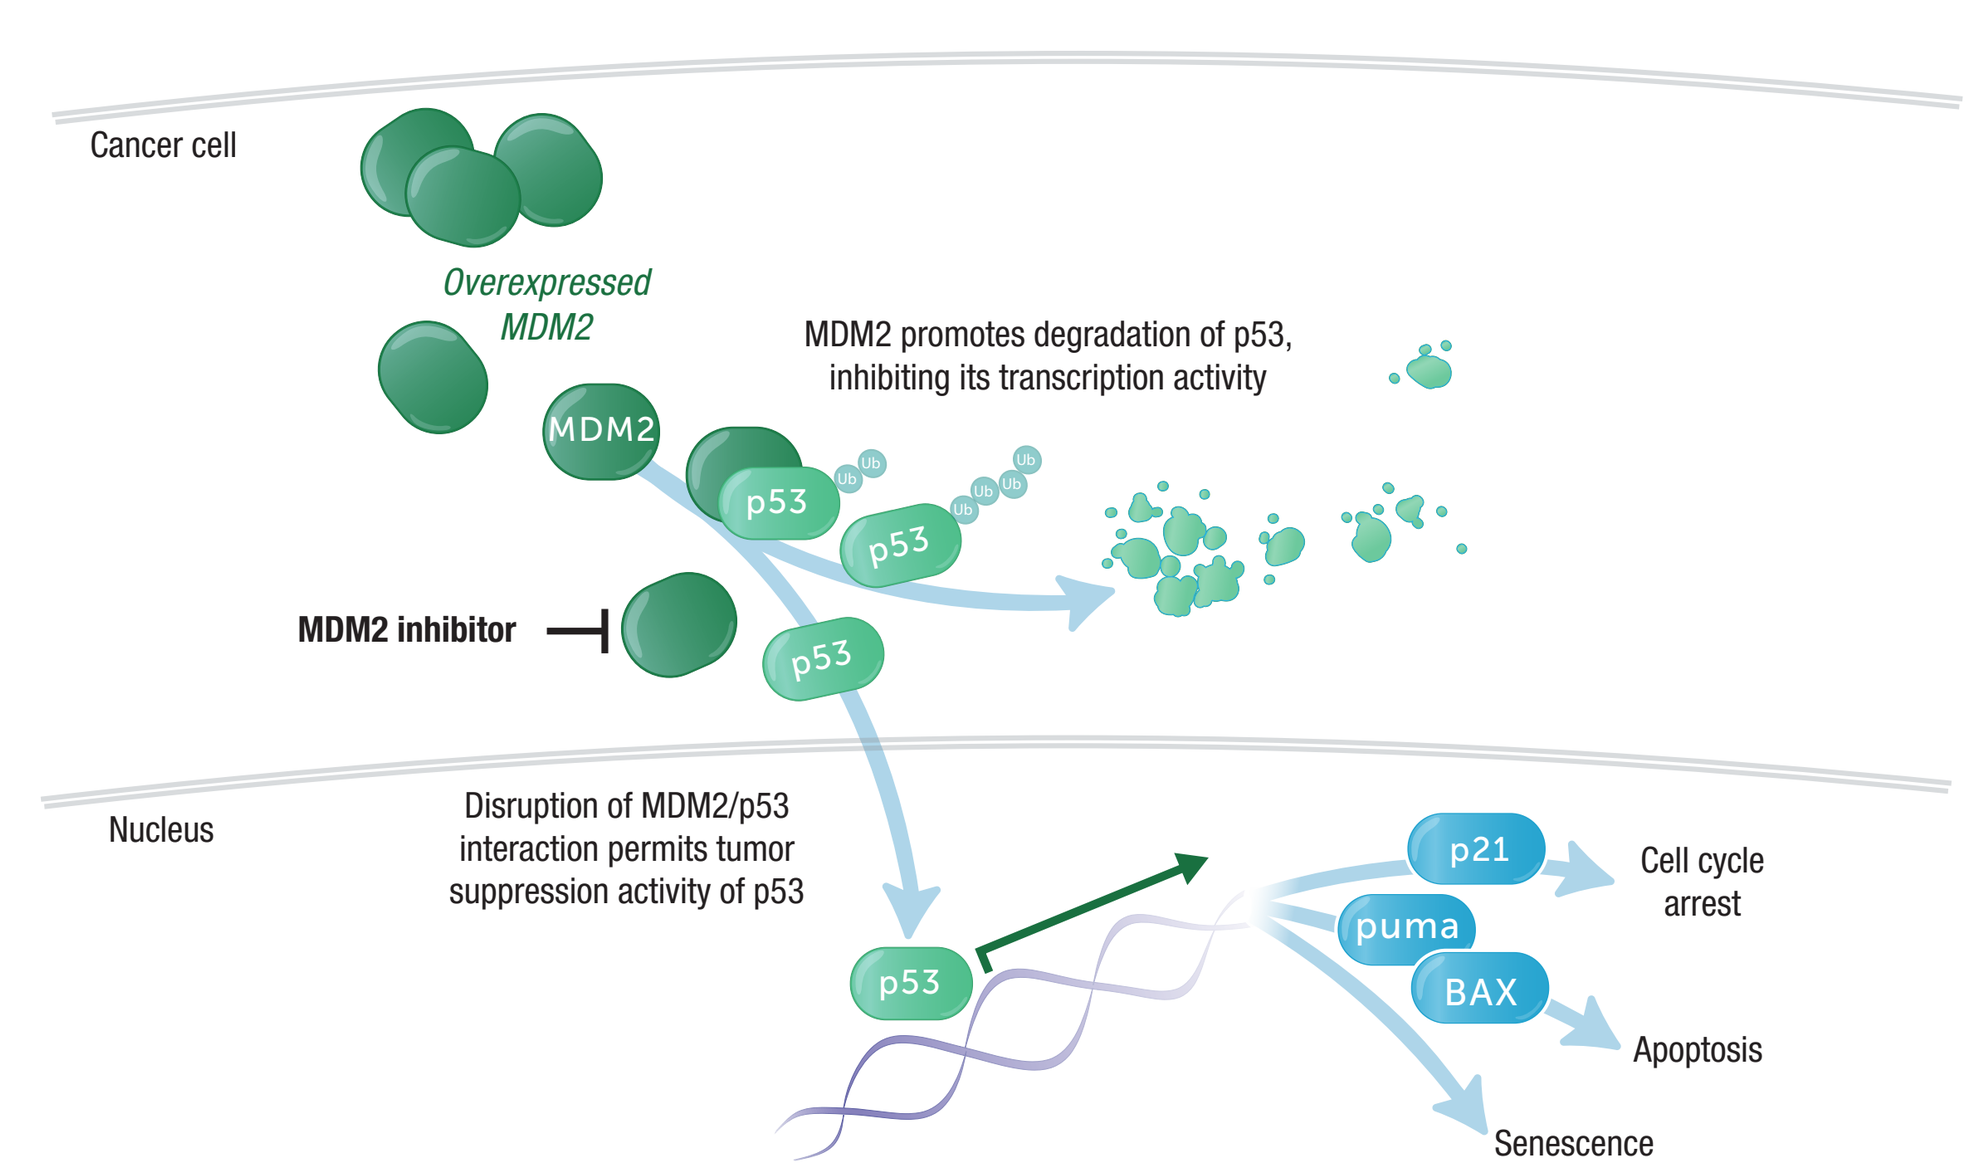
Cancer cell
Overexpressed MDM2
MDM2 promotes degradation of p53, inhibiting its transcription activity
MDM2
p53
MDM2 inhibitor
Nucleus
Disruption of MDM2/p53 interaction permits tumor suppression activity of p53
p21
puma
BAX
Cell cycle arrest
Apoptosis
Senescence

- AZA is a hypomethylating agent that is part of the standard of care for patients with AML and MDS[4,5]
- Reactivation of p53-inducible genes with milademetan combined with hypomethylation and cytotoxicity with AZA has shown increased activity compared with single-agent milademetan or AZA in preclinical models of AML (**Figures 2-4**)[6]

Figure 2. Sequential Treatment (AZA → milademetan) Enhanced AML Growth Inhibition In Vivo Using *TP53* Wild-Type MOLM-13 Cells

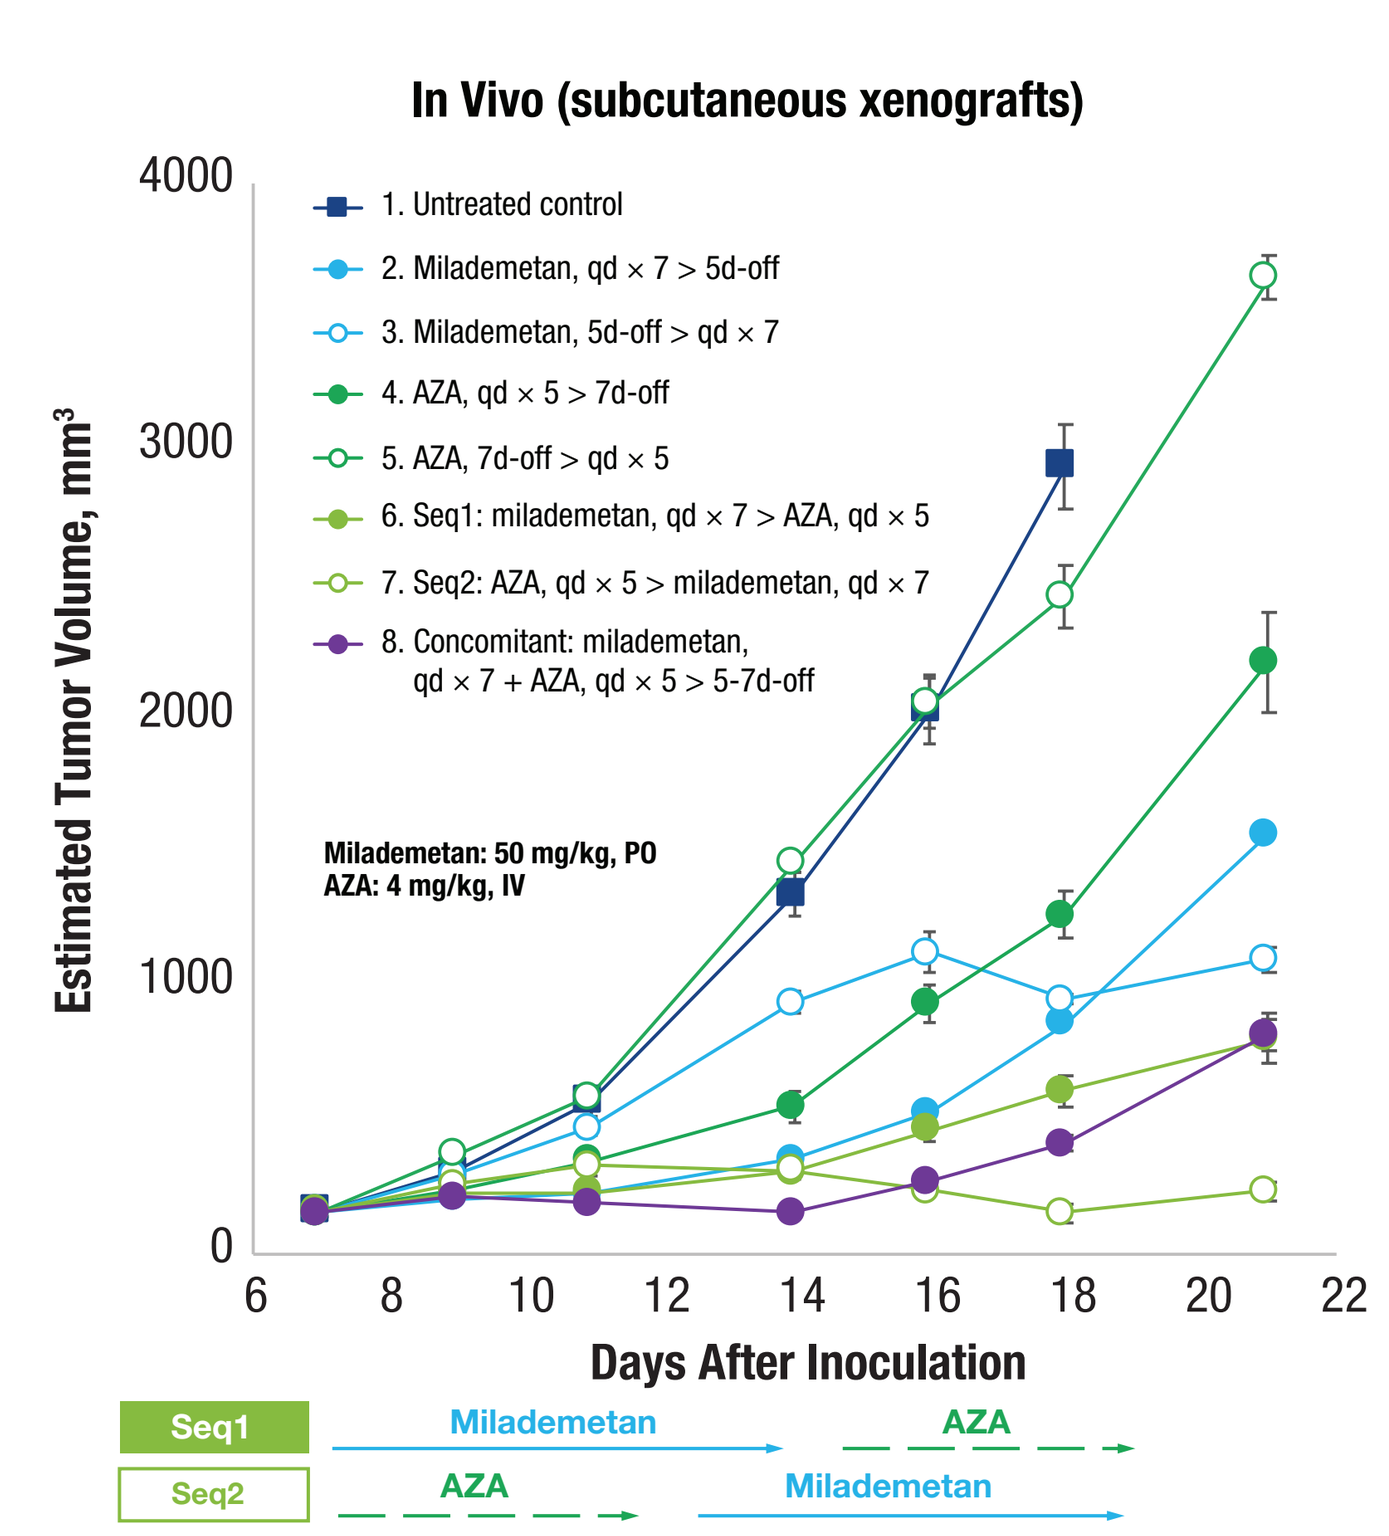
In Vivo (subcutaneous xenografts)
Estimated Tumor Volume, mm³
Days After Inoculation
1. Untreated control
2. Milademetan, qd × 7 > 5d-off
3. Milademetan, 5d-off > qd × 7
4. AZA, qd × 5 > 7d-off
5. AZA, 7d-off > qd × 5
6. Seq1: milademetan, qd × 7 > AZA, qd × 5
7. Seq2: AZA, qd × 5 > milademetan, qd × 7
8. Concomitant: milademetan, qd × 7 + AZA, qd × 5 > 5-7d-off
Milademetan: 50 mg/kg, PO
AZA: 4 mg/kg, IV
Seq1
Seq2
Milademetan
AZA

Each data point and bar represents the mean and SE of the estimated tumor volume in each group, respectively (n = 6). IV, intravenously; PO, orally; qd, once daily; seq, sequential treatment.

Figure 3. AZA Pretreatment Enhanced Milademetan-Induced Growth Inhibitory Effects and Apoptosis in *TP53* Wild-Type AML Cell Lines

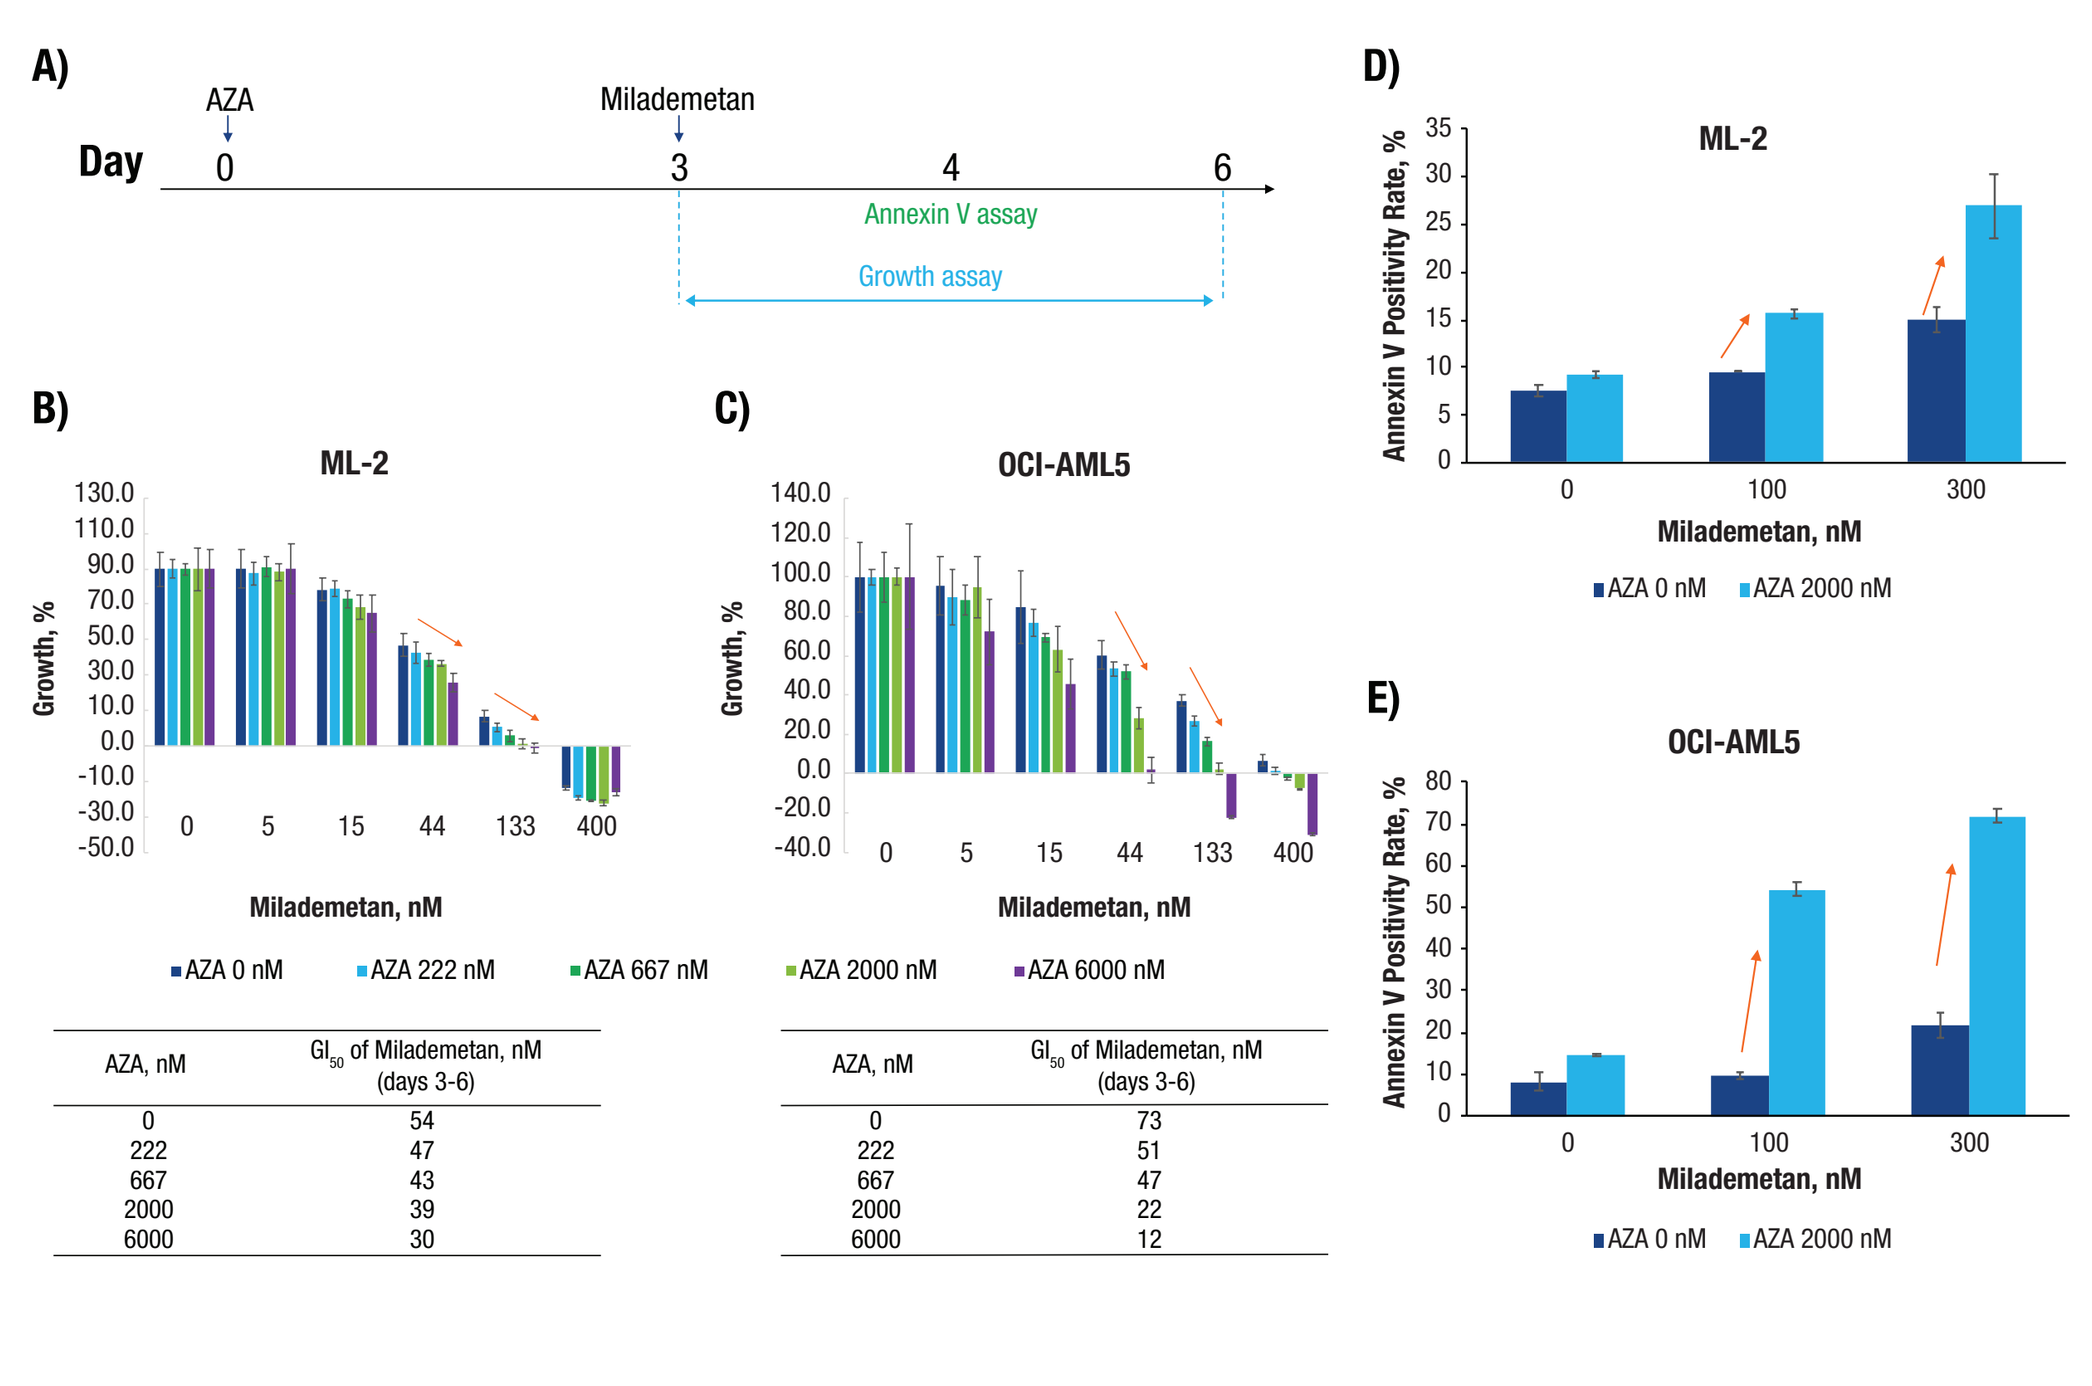
A)
Day 0 3 4 6
AZA
Milademetan
Annexin V assay
Growth assay
B) ML-2
C) OCI-AML5
Growth, %
Milademetan, nM
AZA 0 nM
AZA 222 nM
AZA 667 nM
AZA 2000 nM
AZA 6000 nM
D) ML-2
E) OCI-AML5
Annexin V Positivity Rate, %
AZA 0 nM
AZA 2000 nM

AZA, nM	GI_{50} of Milademetan, nM (days 3-6)
0	54
222	47
667	43
2000	39
6000	30

AZA, nM	GI_{50} of Milademetan, nM (days 3-6)
0	73
222	51
667	47
2000	22
6000	12

Schematic representation of the treatment schedule of AZA and milademetan (A). Cell growth from days 3 to 6 and GI_{50} of milademetan at each concentration of AZA were calculated for ML-2 (B) and OCI-AML5 cells (C) based on ATP assay data. Each bar and error bar represents the mean and SD (n = 4). Annexin V positivity rate was determined for ML-2 (D) and OCI-AML5 cells (E). Each data point and error bar represents the mean and SD (n = 3).

Figure 4. AZA Induced Genome-Wide Demethylation of DNA, Including Promoter Regions of p53 Target Genes Related to Cell Cycle Arrest and Apoptosis

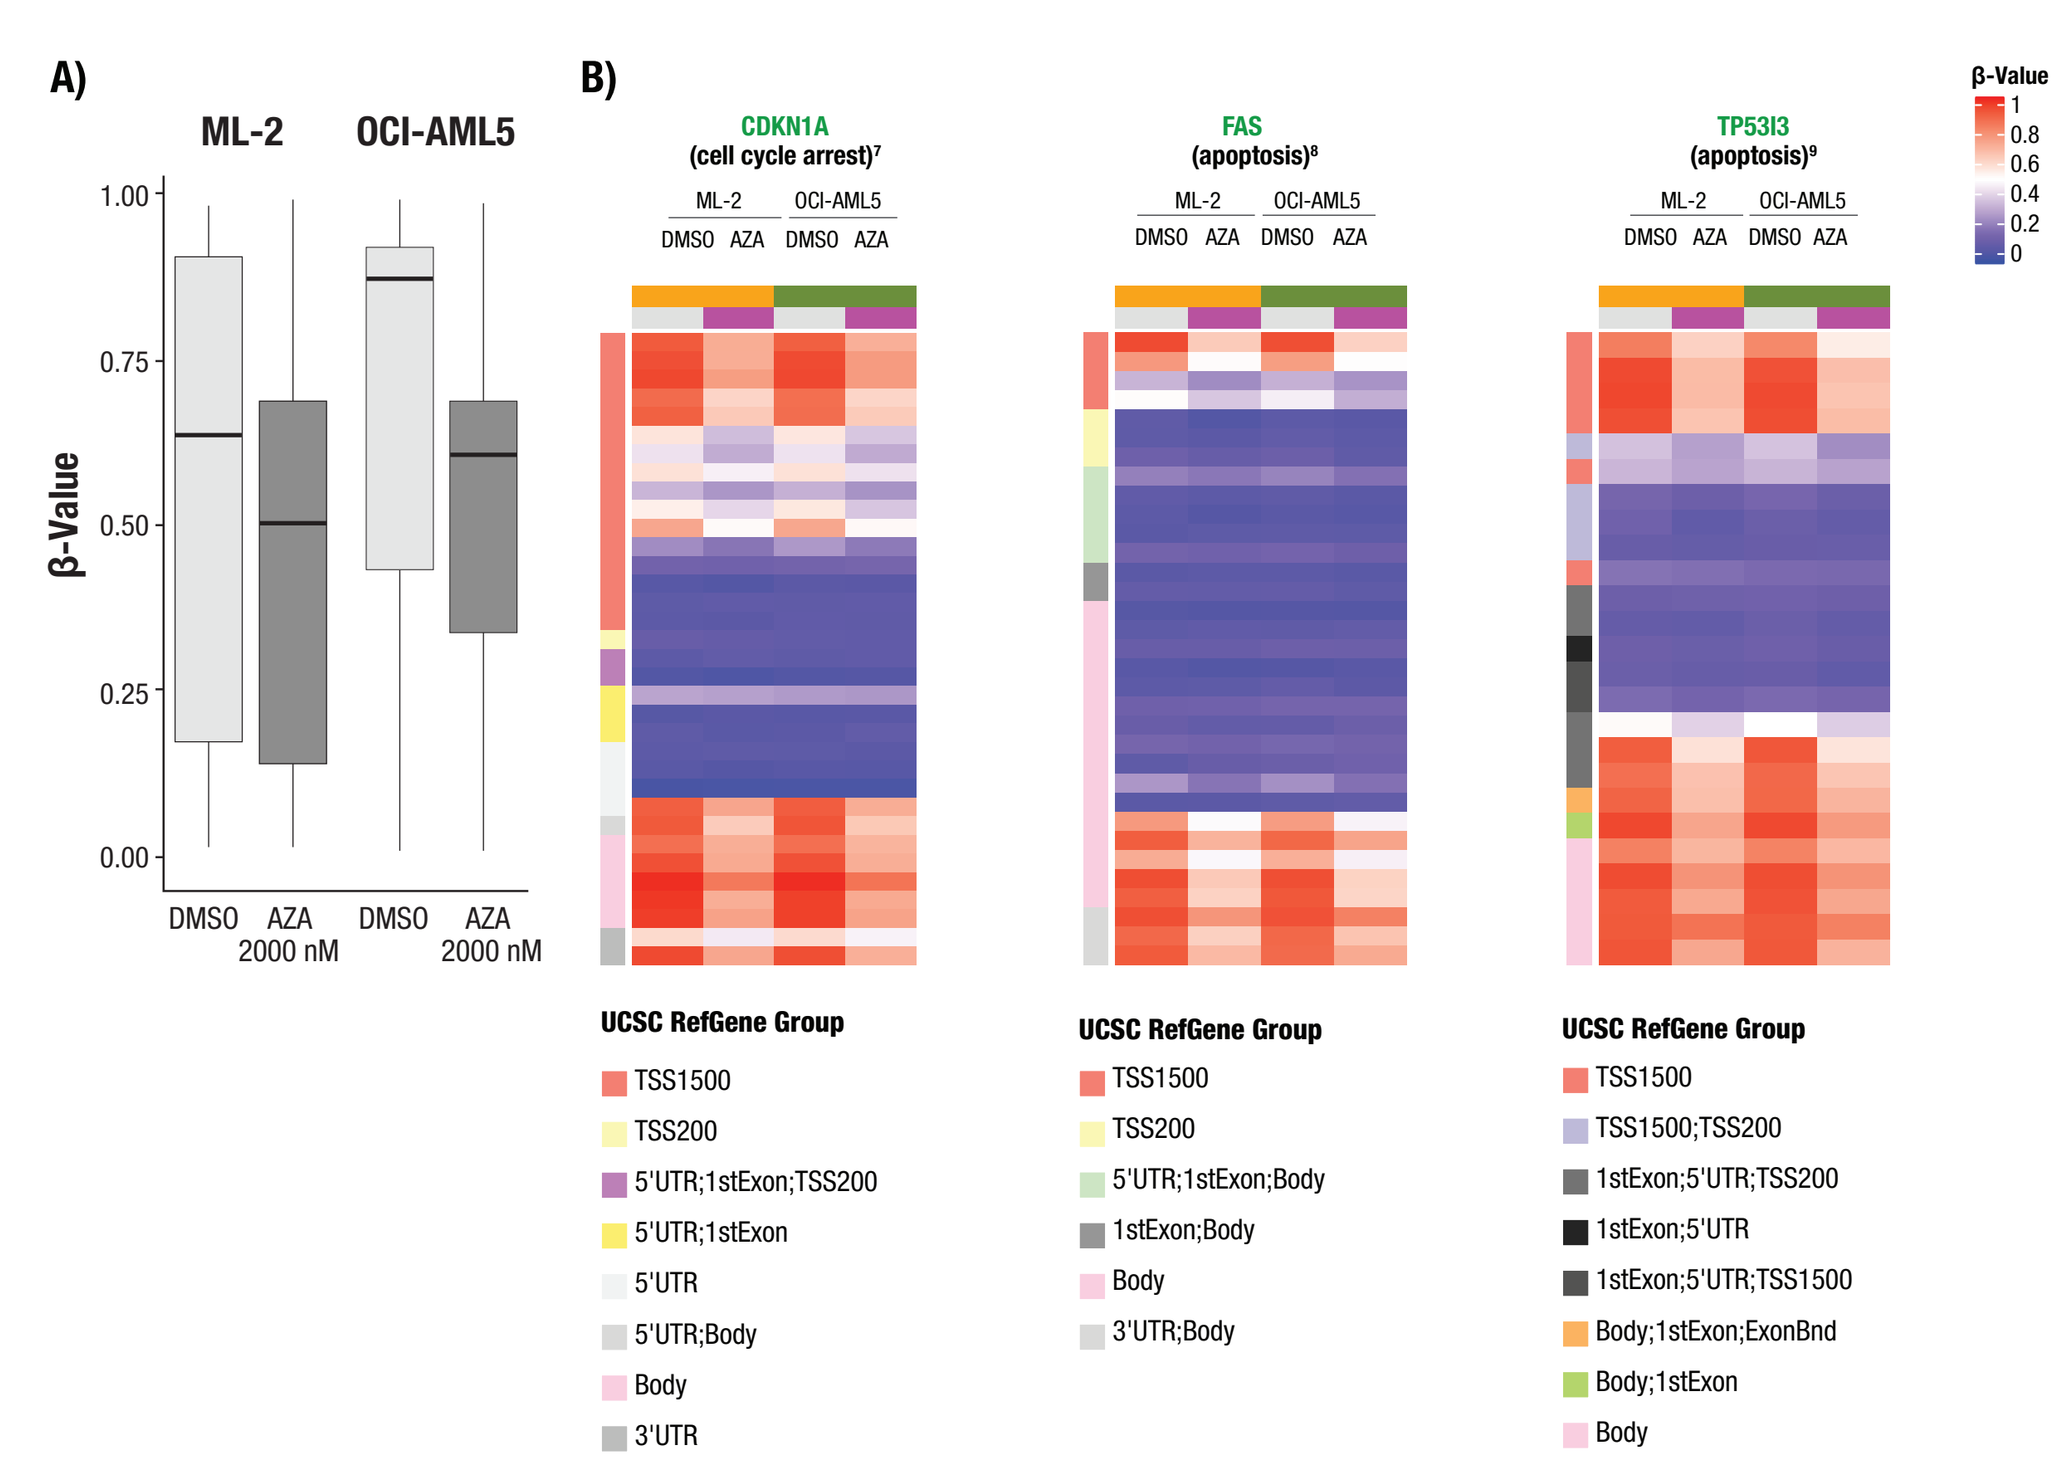
A)
ML-2
OCI-AML5
β-Value
DMSO
AZA 2000 nM
B)
CDKN1A (cell cycle arrest)
FAS (apoptosis)
TP53I3 (apoptosis)
UCSC RefGene Group
TSS1500
TSS200
5'UTR;1stExon;TSS200
5'UTR;1stExon
5'UTR
5'UTR;Body
Body
3'UTR
5'UTR;1stExon;Body
1stExon;Body
3'UTR;Body
TSS1500;TSS200
1stExon;5'UTR;TSS200
1stExon;5'UTR
1stExon;5'UTR;TSS1500
Body;1stExon;ExonBnd
Body;1stExon

(A) DNA methylation levels in ML-2 and OCI-AML5 cells treated with AZA for 3 days. Methylation profile of genome-wide DNA and the p53 target genes cyclin-dependent kinase inhibitor 1A (*CDKN1A*); tumor necrosis factor receptor superfamily, member 6 (*FAS*); and tumor protein p53-inducible protein 3 (*TP53I3*) (B). TSS200, 0-200 bases upstream of the transcriptional start site (TSS); TSS1500, 200-1500 bases upstream of the TSS; 5'UTR, within the 5' untranslated region, between the TSS and the ATG start site; Body, between the ATG and stop codon, irrespective of the presence of introns, exons, TSS, or promoters; 3'UTR, between the stop codon and poly A signal. TSS1500 and TSS200 are categorized as promoter regions.[10]

OBJECTIVES

- The key objectives of this study are to evaluate the safety, tolerability, and preliminary efficacy of and determine the recommended phase 2 dose for the combination of milademetan with AZA in patients with AML or high-risk MDS (**Figure 5**, **Table 1**, and **Table 2**)

METHODS

Study Design

- Open-label phase 1 study with a dose-escalation part (parts 1 and 1A) and dose-expansion part (part 2) (**Figure 5**)
- The expansion part consists of 3 cohorts: adult patients with relapsed/refractory (R/R) AML, newly diagnosed AML unfit for intensive chemotherapy, or high-risk MDS (**Table 1**)
- During dose escalation, milademetan will be administered orally as a single agent (part 1) once daily on days 1 to 21 of a 28-day cycle starting at 60 mg, with proposed levels of 90, 120, 160, and 210 mg (**Figure 5**)
 - In part 1A, AZA will be administered at 75 mg/m² on days 1 to 7 of each 28-day cycle in combination with milademetan treatment on days 5 to 14 or 8 to 14
 - An alternative schedule of AZA at 75 mg/m² on days 1 to 5 and 8 to 9 will also be tested
- The dose-escalation and dose-expansion parts of the study will be conducted at 12 sites in the United States

Figure 5. Study Design

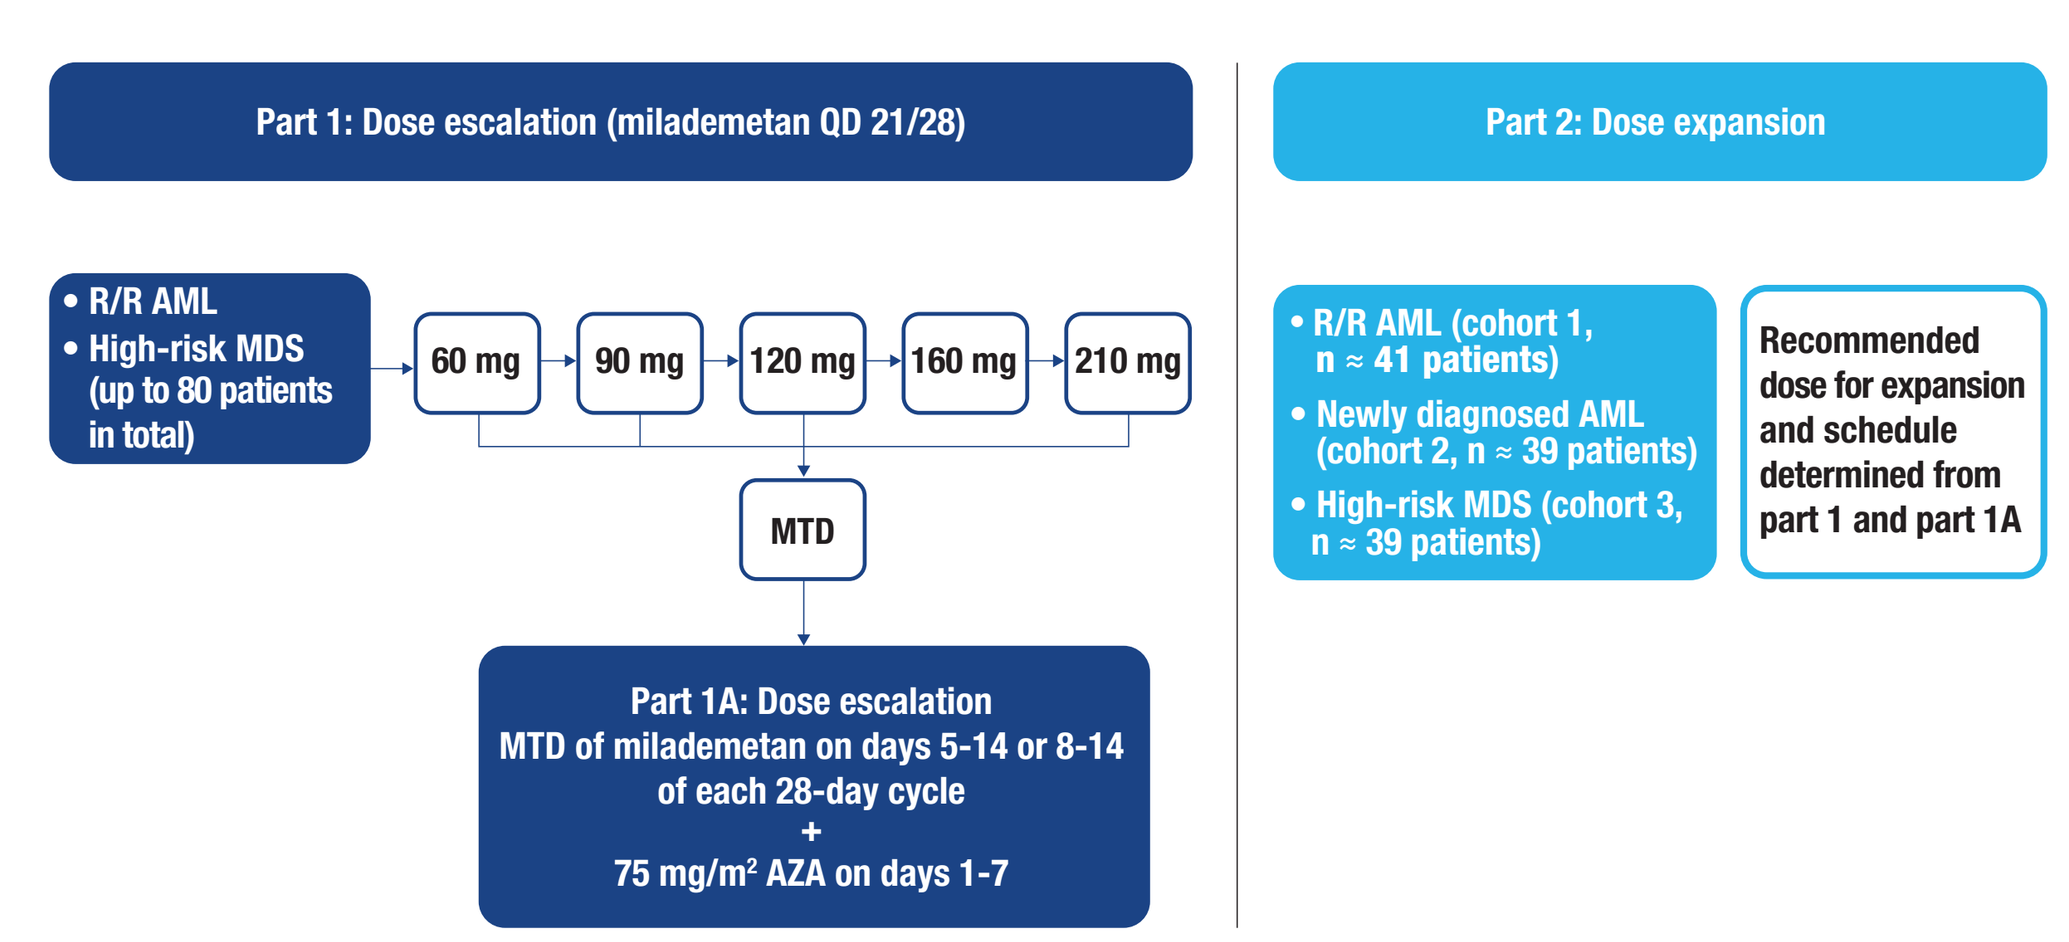
Part 1: Dose escalation (milademetan QD 21/28)
• R/R AML
• High-risk MDS (up to 80 patients in total)
60 mg
90 mg
120 mg
160 mg
210 mg
MTD
Part 1A: Dose escalation
MTD of milademetan on days 5-14 or 8-14 of each 28-day cycle
+
75 mg/m² AZA on days 1-7
Part 2: Dose expansion
• R/R AML (cohort 1, n ≈ 41 patients)
• Newly diagnosed AML (cohort 2, n ≈ 39 patients)
• High-risk MDS (cohort 3, n ≈ 39 patients)
Recommended dose for expansion and schedule determined from part 1 and part 1A

MTD, maximum tolerated dose.

Table 1. Patient Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• R/R AML or high-risk MDS • Age ≥ 18 years • ECOG PS 0-2 • Adequate renal and hepatic function • Willingness to undergo genotyping for *TP53* mutation **Other criteria for part 2 cohort 2:** • Ineligibility for intensive induction chemotherapy due to ≥ 1 of the following criteria: – Age ≥ 75 years with ECOG PS 0-2 – Age 18-74 years and known history of congestive heart failure, ECOG PS 3, or any other comorbidity that the physician judges to be incompatible with conventional chemotherapy	• Acute promyelocytic leukemia • Active CNS leukemia • Malignancy with known non-synonymous mutation, insertion, or deletion in the *TP53* gene determined previously or at screening • Known HIV or active HBV/HCV infection • Unresolved toxicities from previous anticancer therapy • Prior treatment with MDM2 inhibitors • Receipt of HSCT within 60 days of first dose of milademetan or clinically significant GVHD • Concomitant treatment with a strong CYP3A inducer[a] • Prolonged QT interval (> 480 ms for men and women)

CNS, central nervous system; CYP3A, cytochrome P450 3A isozyme; ECOG PS, Eastern Cooperative Oncology Group performance status; GVHD, graft-vs-host disease; HBV, hepatitis B virus; HCV, hepatitis C virus; HIV, human immunodeficiency virus; HSCT, hematopoietic stem cell transplant.

[a] Concomitant strong CYP3A inhibitor is allowed with dose adjustment.

Table 2. Trial Endpoints

Part 1/1A (dose escalation)	Part 2 (dose expansion)
Primary endpoints	
• Safety and tolerability, including SAEs, TEAEs, vital sign measurements, clinical laboratory parameters, ECG parameters, DLTs • MTD of single-dose milademetan and milademetan in combination with AZA • Recommended dose for expansion	• Confirmation of safety and tolerability • Preliminary efficacy, including: – Patients with AML: DOR; rates of CR, CRi, CRh, PR, SD, MLFS, CRc (CR + CRi), and ORR (CRc + MLFS + PR) – Patients with MDS: rates of CR, PR, marrow CR, SD, cytogenetic response, and hematologic improvement • Recommended phase 2 dose
Secondary endpoints	
• Milademetan PK following single and multiple dosing	• Milademetan PK at the RDE
Exploratory endpoints	
• Treatment responses, biomarker assessments, other PD effects • DDI of milademetan with AZA	• PD effects

CR, complete remission; CRc, composite complete remission; CRh, CR with partial hematologic recovery; CRi, CR with incomplete blood count recovery; DDI, drug-drug interaction; DLT, dose-limiting toxicity; DOR, duration of CRc; ECG, electrocardiogram; MLFS, morphologic leukemia-free state; ORR, objective response rate; PD, pharmacodynamics; PK, pharmacokinetics; PR, partial remission; RDE, recommended dose for expansion; SAE, serious adverse event; SD, stable disease; TEAE, treatment-emergent adverse event.

STATISTICAL DESIGN

- The primary statistical analysis will occur after all patients have discontinued the study or completed ≥ 6 months of treatment
- For response rate, point estimates and 2-sided 95% exact binomial CIs will be calculated
- The time-to-event endpoint (DOR) will be summarized using the Kaplan-Meier method
- PK parameters will be calculated using noncompartmental analysis
- PD effects will be summarized using descriptive statistics

ENROLLMENT

- This trial is currently recruiting patients and is registered at ClinicalTrials.gov (NCT02319369)

REFERENCES

1. Moll UM, et al. *Mol Cancer Res*. 2003;1(14):1001-1008.
2. Carvajal LA, et al. *Sci Transl Med*. 2018;10(436):eaao3003.
3. Ishizawa J, et al. *Cancer Res*. 2018;78(10):2721-2731.
4. Schuh AC, et al. *Crit Rev Oncol Hematol*. 2017;116:159-177.
5. Steensma DP. *Blood Cancer J*. 2018;8(5):47.
6. Noguchi S, et al. *HemaSphere*. 2019;3:65.
7. Harper JW, et al. *Cell*. 1993;75:805-816.
8. Rakkar AN, et al. *Cell Death Differ*. 1999;6:326-333.
9. Porté S, et al. *J Biol Chem*. 2009;284:17194-17205.
10. Bibikova M, et al. *Genomics*. 2011;98:288-295

DISCLOSURES

C. D. DiNardo—Membership on an Entity's Board of Directors or Advisory Committees: Notable Labs; Honoraria & Research Funding: AbbVie, Agios, Celgene, Daiichi Sankyo, Inc.; Honoraria: Jazz Pharmaceuticals, MedImmune, Syros. **R. Olin**—Consultancy & Research Funding: Genentech; Consultancy: Jazz Pharmaceuticals, Revolution Medicine; Research Funding: Astellas, AstraZeneca, Clovis, Daiichi Sankyo, Inc., Ignyta, Mirati Therapeutics, Novartis, Spectrum. **J. Ishizawa**—Patents Licensed & Royalty Bearing: Daiichi Sankyo, Inc. **H. Sumi, J. Xie, P. Kumar**—Employees of Daiichi Sankyo, Inc. **K. Kato**—Employee of Daiichi Sankyo, Inc. and Celgene. **M. Andreeff**—Consultancy: AstraZeneca, Celgene, Jazz Pharmaceuticals, 6 Dimensions Capital; Research Funding: Breast Cancer Research Foundation, Cancer Prevention and Research Institute of Texas, National Institutes of Health/National Cancer Institute (NCI); Equity Ownership: Aptose, Eutropics, Oncoceutics, Oncolyze, Reata; Membership on an Entity's Board of Directors or Advisory Committees: BioLineRx, Cancer UK, Center for Drug Research & Development, Chronic Lymphocytic Leukemia Foundation, German Research Council, Leukemia & Lymphoma Society, NCI Cancer Therapy Evaluation Program, NCI Rare Diseases Clinical Research Network; Equity Ownership & Membership on an Entity's Board of Directors or Advisory Committees: Senti Bio; Consultancy, Patents, & Royalties (patents licensed, royalty bearing, and research funding): Daiichi Sankyo, Inc.

ACKNOWLEDGMENTS

This study is sponsored by Daiichi Sankyo and conducted in collaboration with The University of Texas MD Anderson Cancer Center. The authors thank the patients and their families for participating in this study. Medical writing support was provided by Jamie King, PhD, of SciMentum, Inc, a Nucleus Global company, and was funded by Daiichi Sankyo, Inc. Editorial support was provided in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

To download a copy of this poster:
Scan QR code via a barcode reader application

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASH and the authors of this poster.

ASH Annual Meeting; December 7-10, 2019; Orlando, Florida.