European Society for Medical Oncology Congress 2019 | September 27-October 1, 2019 | Barcelona, Spain

Poster 885P

Spatial-Temporal Change in Quantitative Total Bone Imaging and Circulating Tumor Cells in Metastatic Castration-Resistant Prostate Cancer Treated With Enzalutamide

Glenn Liu,[1,2] Christos E. Kyriakopoulos,[1] Joshua M. Lang,[1] Jamie M. Sperger,[1] Anupama Singh,[1] Elisabeth I. Heath,[3] Scott Perlman,[1] Tina Mayer,[4] Katharina Modelska,[5] Anthony Porcari,[6] William Duggan,[7] Robert Jeraj[1,2]

[1]University of Wisconsin Carbone Cancer Center, University of Wisconsin, Madison, WI, USA; [2]AIQ Solutions, Madison, WI, USA; [3]Karmanos Cancer Institute, Wayne State University School of Medicine, Detroit, MI, USA;
[4]Cancer Institute of New Jersey, Rutgers University, New Brunswick, NJ, USA; [5]Pfizer Inc., San Francisco, CA, USA; [6]Pfizer Inc., Collegeville, PA, USA; [7]Pfizer Inc., Groton, CT, USA

BACKGROUND

- Cancer is a dynamic disease which, over time, may result in nonuniform distribution of distinct tumor cell subpopulations at different sites (spatial heterogeneity) or an increase in molecularly diverse cells (temporal heterogeneity).[1]
- Quantitative total bone imaging (QTBI) is an advanced analytics platform that automatically localizes, identifies, quantitates, and matches osteoblastic metastases within an individual to provide spatio-temporal lesion-level treatment response information from ^{18}F-sodium fluoride (^{18}F-NaF) positron emission tomography/computed tomography (PET/CT) scans (**Figure 1**).
- Vertical exclusion-based rare sample analysis (VERSA) is a device that allows circulating tumor cell (CTC) capture and purification for protein and gene expression analysis from peripheral blood (**Figure 2**).
- We have previously reported the primary results of our study showing that all patients with metastatic castration-resistant prostate cancer (mCRPC) to bone treated with enzalutamide had at least one responding lesion at the time of prostate-specific antigen (PSA), standard radiographic, or clinical progression or at 2 years without progression.[2]
- Here we report the exploratory objectives of tumor burden and CTC analysis using QTBI and VERSA in men with mCRPC to bone treated with enzalutamide.

Figure 1. Examples of QTBI

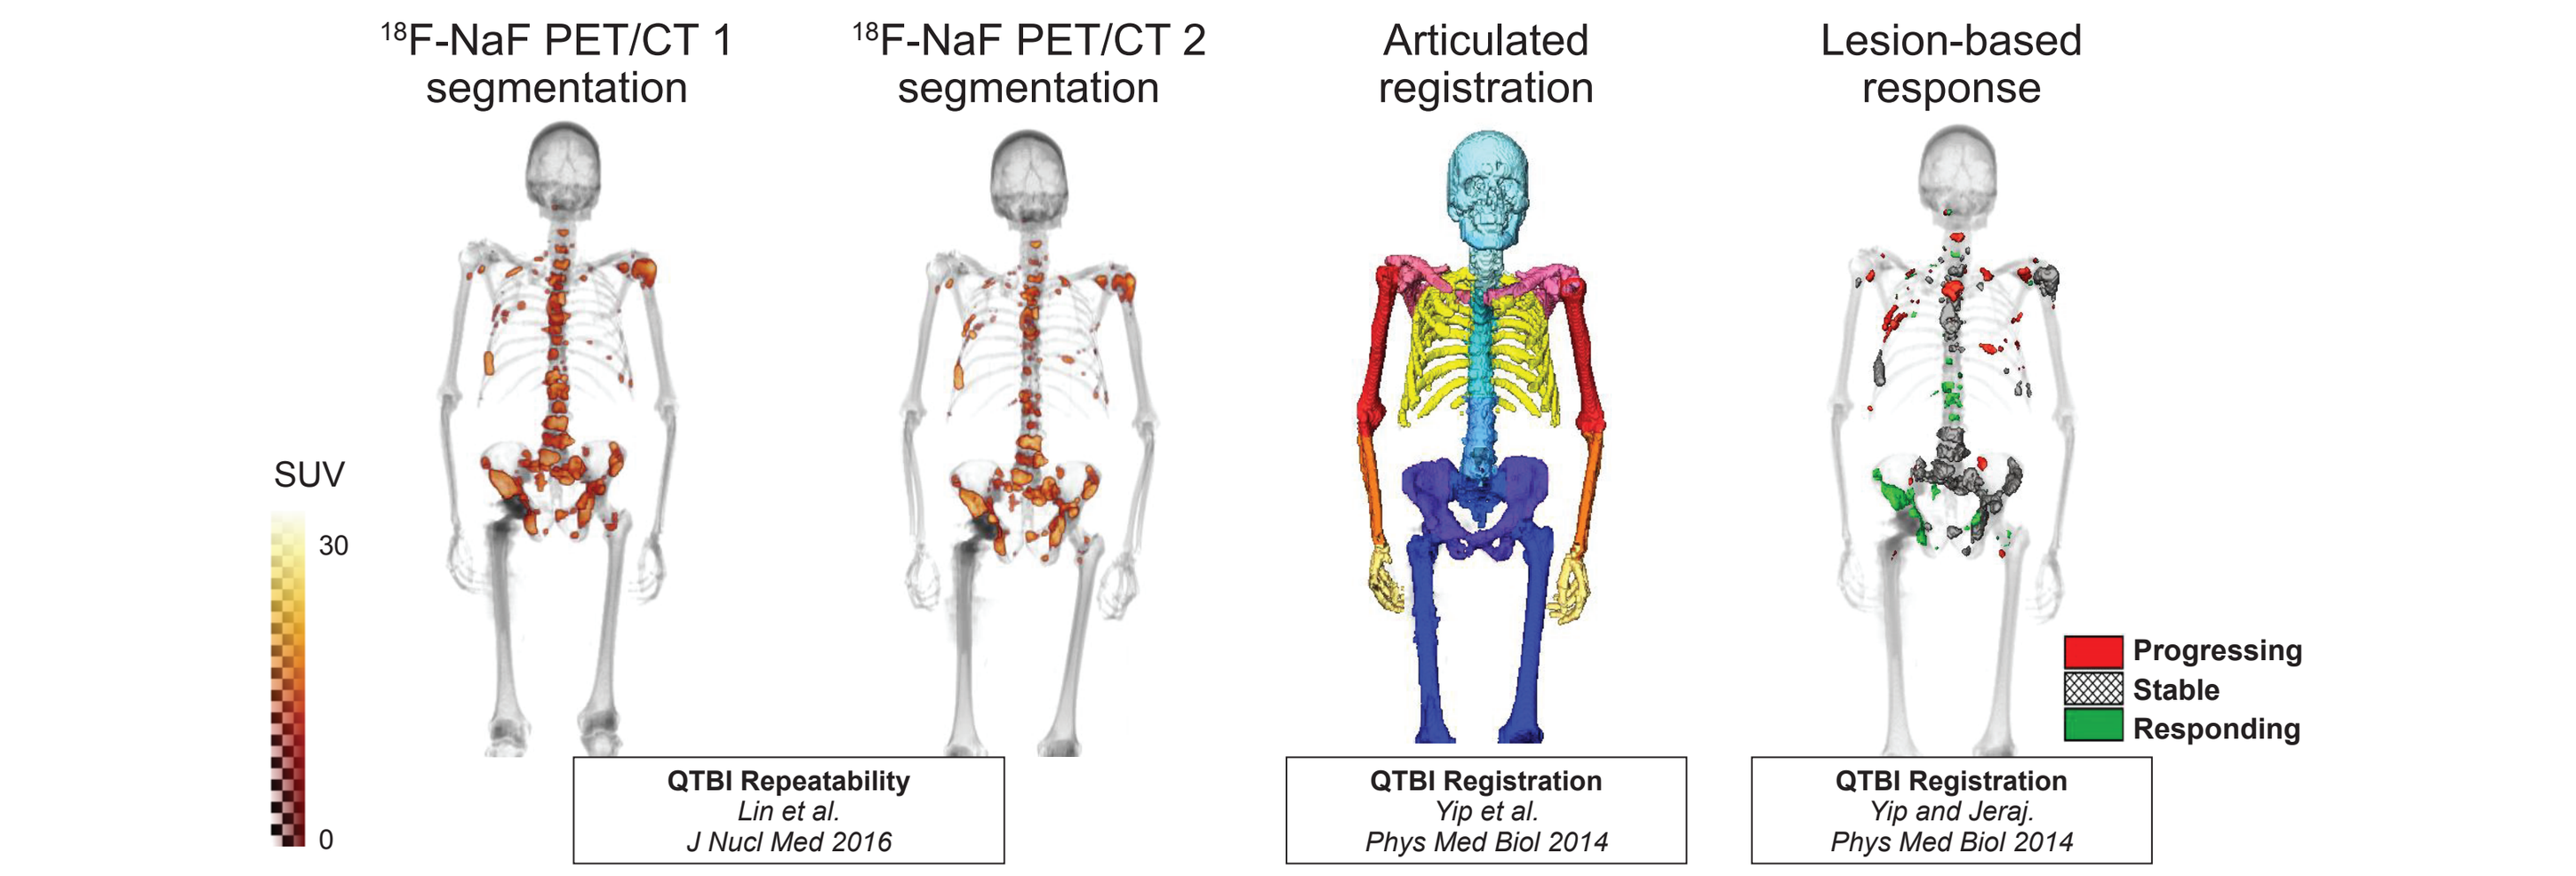
^{18}F-NaF PET/CT 1 segmentation
^{18}F-NaF PET/CT 2 segmentation
Articulated registration
Lesion-based response
SUV
30
0
Progressing
Stable
Responding
QTBI Repeatability Lin et al. J Nucl Med 2016
QTBI Registration Yip et al. Phys Med Biol 2014
QTBI Registration Yip and Jeraj. Phys Med Biol 2014

QTBI-identified lesions on PET/CT are automatically segmented. Using articulated registration, the skeleton can be aligned precisely, allowing for lesion-based treatment response assessment.
Abbreviations: ^{18}F-NaF, ^{18}F-sodium fluoride; PET/CT, positron emission tomography/computed tomography; QTBI, quantitative total bone imaging; SUV, standardized uptake value

Figure 2. The VERSA Platform

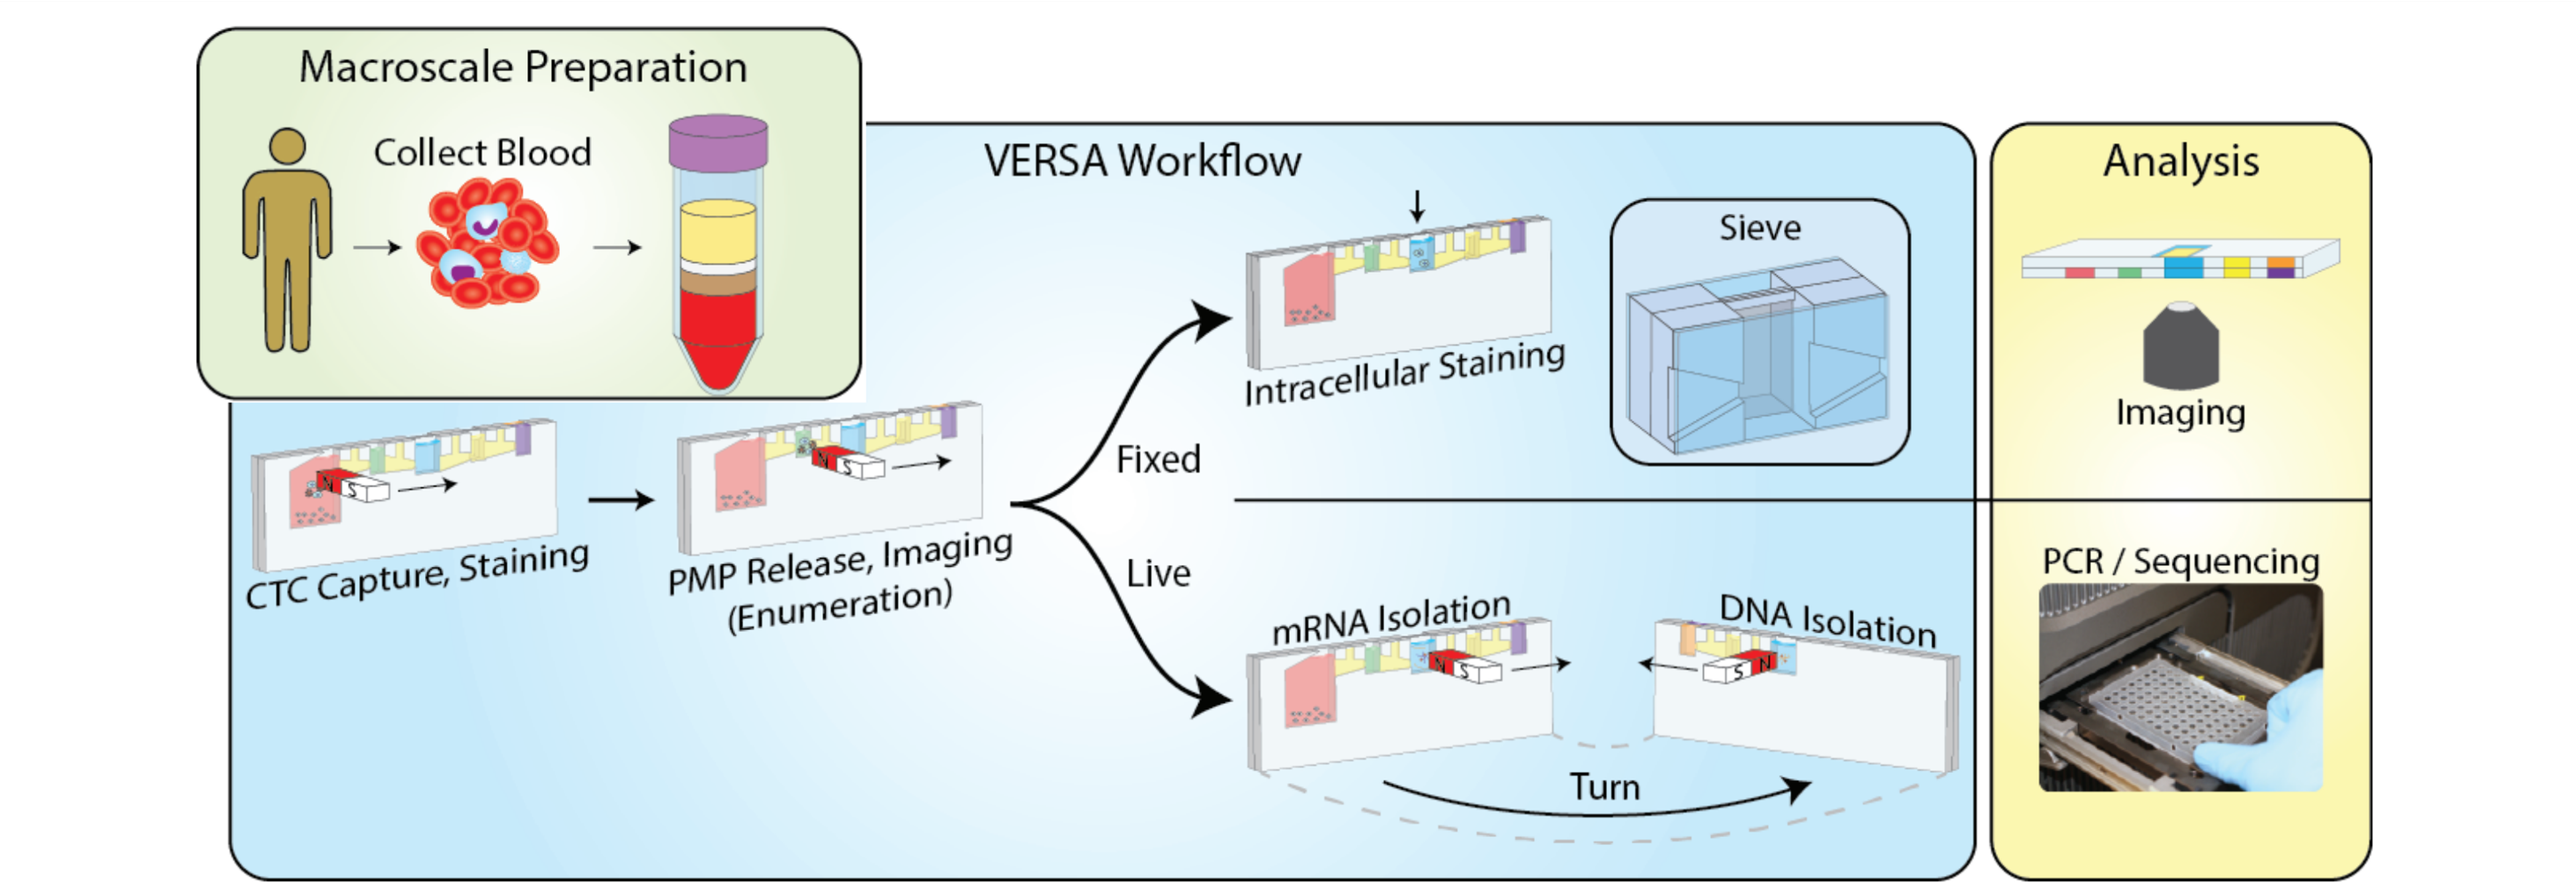
Macroscale Preparation
Collect Blood
VERSA Workflow
Sieve
Intracellular Staining
Fixed
Live
CTC Capture, Staining
PMP Release, Imaging (Enumeration)
mRNA Isolation
DNA Isolation
Turn
Analysis
Imaging
PCR / Sequencing

The VERSA platform integrates efficient cell capture with PMP removal, a staining well (sieve), and a downstream method to capture both mRNA and DNA without dilution steps. Endpoints enabled in VERSA allow for multiple analyte interrogation in an integrated and flexible workflow.
Abbreviations: CTC, circulating tumor cell; PCR, polymerase chain reaction; PMP, paramagnetic particle; VERSA, vertical exclusion-based rare sample analysis.

METHODS

- **Figure 3** illustrates the study schema.

Figure 3. Study Schema

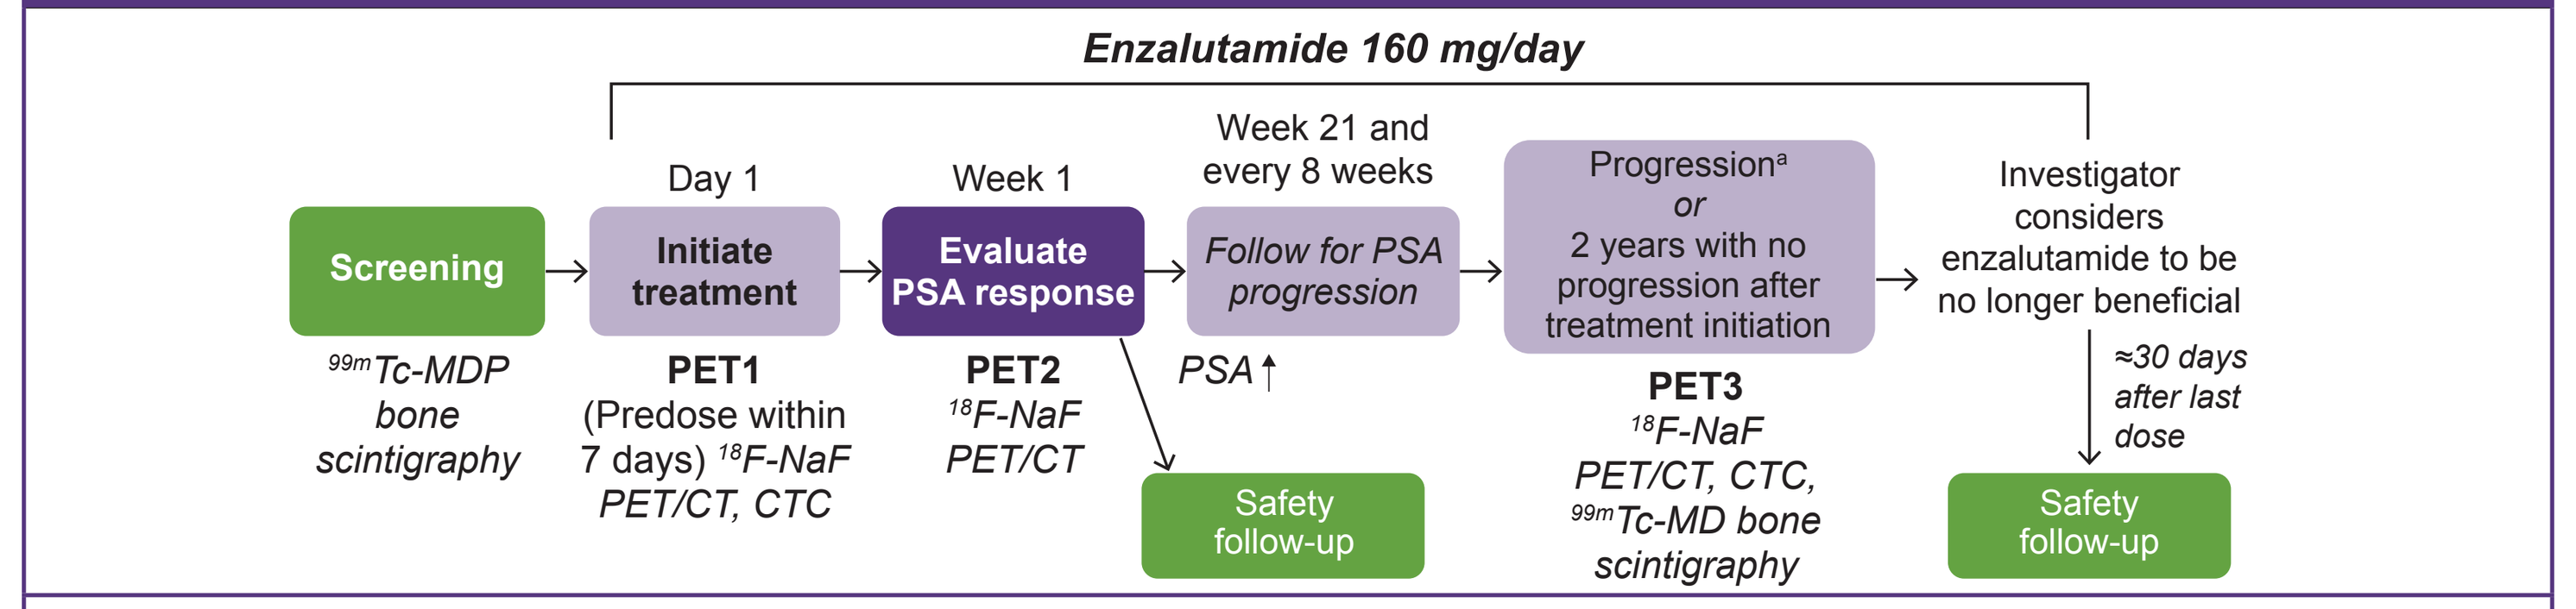
Enzalutamide 160 mg/day
Screening
^{99m}Tc-MDP bone scintigraphy
Day 1
Initiate treatment
PET1 (Predose within 7 days) ^{18}F-NaF PET/CT, CTC
Week 1
Evaluate PSA response
PET2 ^{18}F-NaF PET/CT
Week 21 and every 8 weeks
Follow for PSA progression
PSA↑
Safety follow-up
Progression[a] or 2 years with no progression after treatment initiation
PET3 ^{18}F-NaF PET/CT, CTC, ^{99m}Tc-MD bone scintigraphy
Investigator considers enzalutamide to be no longer beneficial
≈30 days after last dose
Safety follow-up

[a]PSA, standard radiographic (bone or soft tissue), or clinically relevant (eg, significant bone pain or skeletal-related event) progression. Patients who did not progress on enzalutamide and were considered by the investigator to benefit from treatment had the option to continue enzalutamide.
Abbreviations: ^{18}F-NaF, ^{18}F-sodium fluoride; ^{99m}Tc-MDP, technetium-99m-methylene diphosphonate; CTC, circulating tumor cell; PET1, baseline; PET2, week 13; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression; PET/CT, positron emission tomography/computed tomography; PSA, prostate-specific antigen.

- ^{18}F-NaF PET/CT scans were obtained at baseline (PET1), week 13 (PET2), and at the time of PSA progression (defined per Prostate Cancer Working Group 2 guidelines as an increase of ≥ 25% and ≥ 2.0 ng/mL above nadir[3]), standard radiographic or clinical progression or at 2 years without progression (PET3) based on QTBI.
- CTCs were obtained at baseline and at PET3.

Objectives and Endpoints

- The primary endpoint was the proportion of men with ≥ 1 responding bone lesion at PET3 (defined as a lesion with a total ^{18}F-NaF standardized uptake value [SUV_{total}] less than baseline).
 - Evaluable men had scans at PET1 and PET3.
- Exploratory objectives were to:
 - Evaluate change in total tumor burden (SUV_{total})
 - Evaluate change in heterogeneity of response across all bone lesions, as measured by SUV_{hetero}
 - Correlate various ^{18}F-NaF uptake measures with efficacy measures
 - Evaluate epithelial cell adhesion molecule (EpCAM)-positive CTCs
 - Correlate CTC analysis with efficacy measures

Eligibility Criteria

- Histologically or cytologically confirmed adenocarcinoma of the prostate without neuroendocrine differentiation or signet-cell or small-cell features
- Presence of bone metastatic disease, as assessed by ≥ 2 lesions on whole-body radionuclide ^{99m}Tc-MDP bone scintigraphy
- Progressive disease on androgen deprivation therapy at screening, defined as a minimum of 2 sequentially rising PSA values, with the last PSA value at ≥ 2 ng/mL or prior bilateral orchiectomy
- No prior enzalutamide, abiraterone acetate, aminoglutethimide, ketoconazole, radium-223 dichloride (or other bone-targeting radionuclides), cytotoxic chemotherapy, or investigational agents that inhibit the androgen receptor or androgen synthesis for castration-resistant prostate cancer
- No visceral metastasis; adenopathy allowed
- No radiation therapy to bone within 4 weeks of enrollment

RESULTS

Demographic and Baseline Characteristics

- A total of 23 men with a median age of 72 years (range, 51-93) and a median PSA of 20.5 ng/mL (range, 3.9-133.6) were enrolled (**Table 1**).

Table 1. Demographic and Baseline Patient Characteristics

Characteristic	ENZA (N = 23)
Age, median (range), y	72 (51-93)
Race, white, no. (%)	21 (91.3)
ECOG PS, no. (%)	
0	16 (69.6)
1	7 (30.4)
Serum PSA, median (range), ng/mL	20.5 (3.9-133.6)
Gleason score, median (range)	7.0 (5.0-9.0)
Prior radiotherapy, no. (%)	
Yes	14 (60.9)
No	9 (39.1)
Prior surgery, no. (%)	
Yes	15 (65.2)
No	8 (34.8)

Abbreviations: ECOG PS, Eastern Cooperative Oncology Group performance status; ENZA, enzalutamide; PSA, prostate-specific antigen.

Primary Endpoint

- The study met its primary endpoint, with 22 of 22 (100%) evaluable men having ≥ 1 responding bone lesion on QTBI at PET3.[2]

Tumor Burden and Heterogeneity

- The mean number of bone lesions at PET1 was 58 (range, 9-168).
- SUV_{total} did not change significantly from baseline to PET3 (least squares [LS] mean percent change from baseline, -3.4%; 95% CI, -35.7 to 45.4).
- SUV_{hetero} was decreased at PET3 compared with baseline (LS mean percent change from baseline, -24.4%; 95% CI, -44.2 to 2.4).
- Examples of individual patient responses are provided in **Figure 4**.

Figure 4. Examples of Patients With (A) Good Response and (B) Suboptimal Response to Enzalutamide

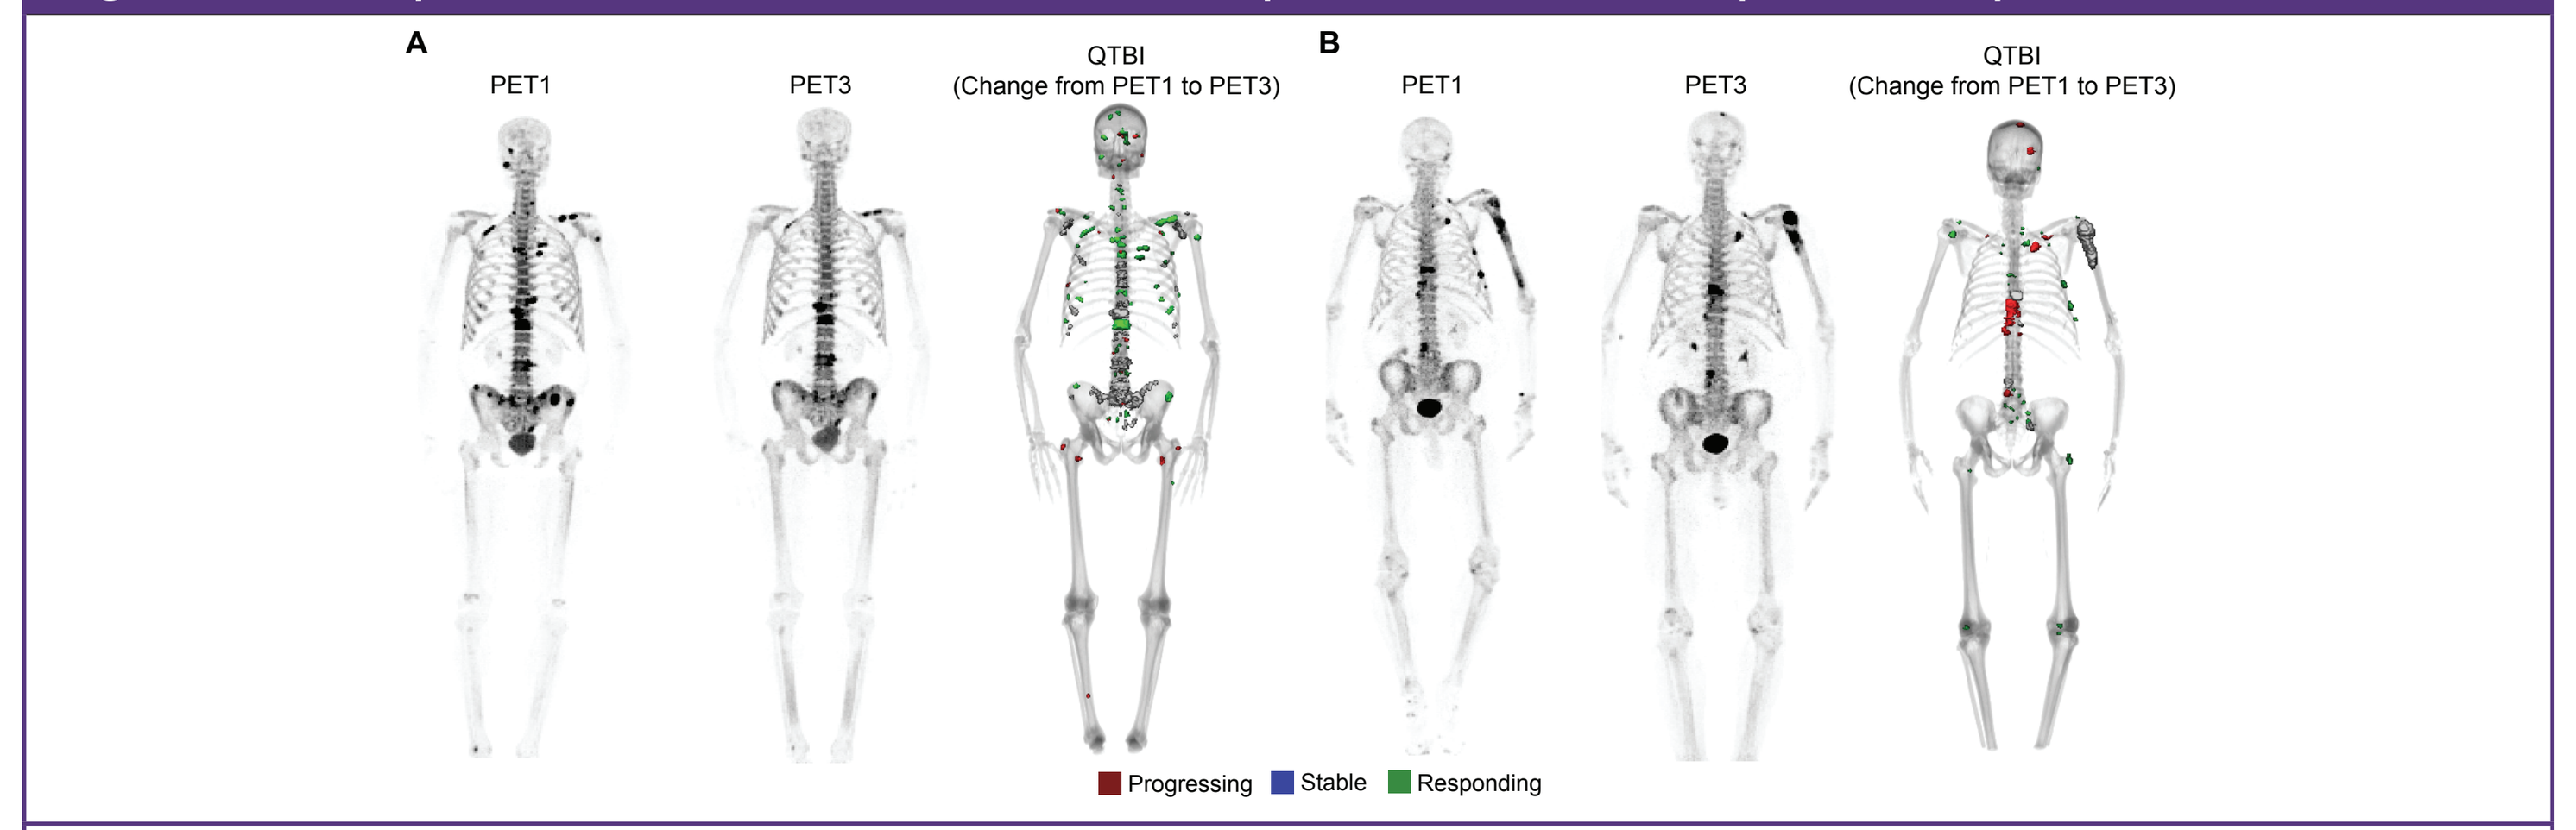
A
PET1
PET3
QTBI (Change from PET1 to PET3)
B
PET1
PET3
QTBI (Change from PET1 to PET3)
Progressing
Stable
Responding

Abbreviations: PET, positron emission tomography; PET1, baseline; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression; QTBI, quantitative total bone imaging.

Correlation With Efficacy Measures

- Maximal PSA decline was correlated with the change in SUV_{max} (representing the activity of the most aggressive lesion) and SUV_{hetero} at PET2 from baseline (**Figure 5**).

Figure 5. Change From Baseline to PET2 in (A) SUV_{max} and (B) SUV_{hetero}

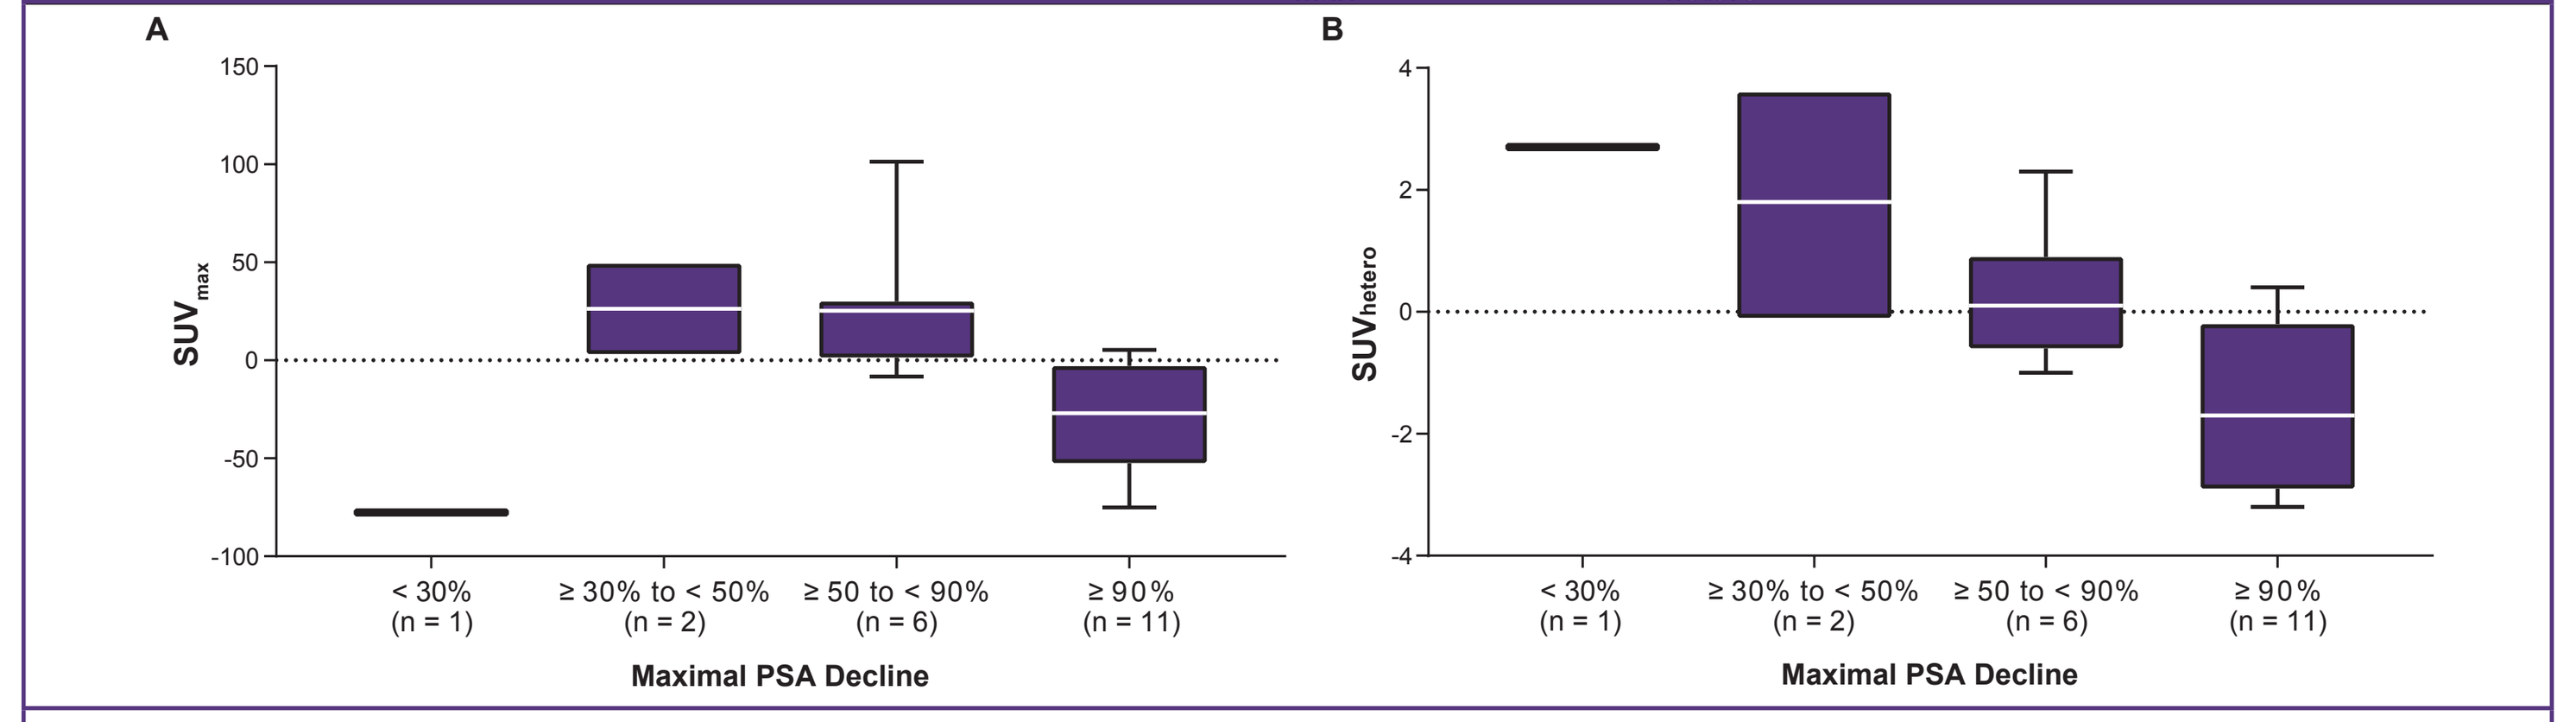
A
SUV_{max}
150
100
50
0
-50
-100
< 30% (n = 1)
≥ 30% to < 50% (n = 2)
≥ 50 to < 90% (n = 6)
≥ 90% (n = 11)
Maximal PSA Decline
B
SUV_{hetero}
4
2
0
-2
-4

Abbreviations: PET, positron emission tomography; PET2, week 13; PSA, prostate-specific antigen; SUV, standardized uptake value; SUV_{hetero}, heterogeneity of response across all bone lesions; SUV_{max}, response in the most aggressive lesion.

- Median duration of treatment was 15.8 months (range, 1.4-34.1).
- Change in lesion heterogeneity (PET1 to PET3) was the most significant predictor of time to PSA progression and time to treatment discontinuation (**Figure 6** and **Figure 7**).

Figure 6. Time to (A) PSA Progression and (B) Treatment Discontinuation by SUV at PET2 and PET3

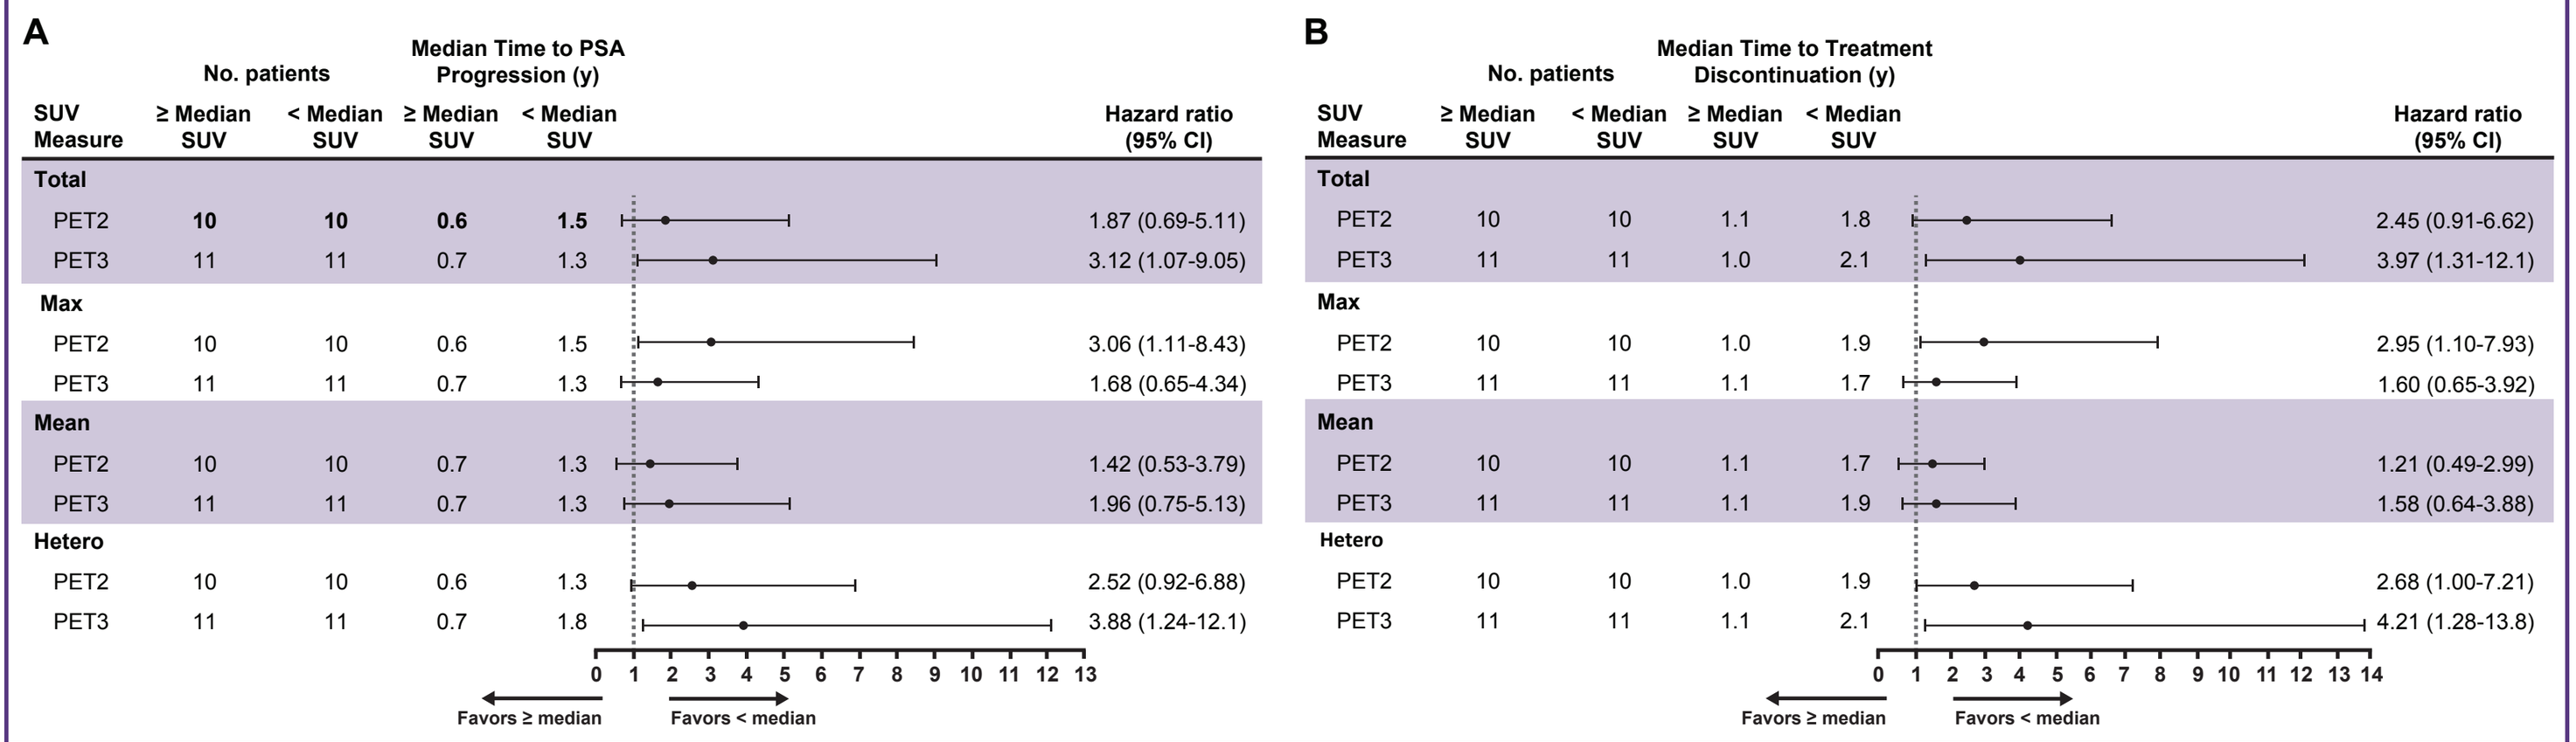
A
SUV Measure | No. patients ≥ Median SUV | < Median SUV | Median Time to PSA Progression (y) ≥ Median SUV | < Median SUV | Hazard ratio (95% CI)
Total
PET2 10 10 0.6 1.5 1.87 (0.69-5.11)
PET3 11 11 0.7 1.3 3.12 (1.07-9.05)
Max
PET2 10 10 0.6 1.5 3.06 (1.11-8.43)
PET3 11 11 0.7 1.3 1.68 (0.65-4.34)
Mean
PET2 10 10 0.7 1.3 1.42 (0.53-3.79)
PET3 11 11 0.7 1.3 1.96 (0.75-5.13)
Hetero
PET2 10 10 0.6 1.3 2.52 (0.92-6.88)
PET3 11 11 0.7 1.8 3.88 (1.24-12.1)
Favors ≥ median
Favors < median
B
SUV Measure | No. patients ≥ Median SUV | < Median SUV | Median Time to Treatment Discontinuation (y) ≥ Median SUV | < Median SUV | Hazard ratio (95% CI)
Total
PET2 10 10 1.1 1.8 2.45 (0.91-6.62)
PET3 11 11 1.0 2.1 3.97 (1.31-12.1)
Max
PET2 10 10 1.0 1.9 2.95 (1.10-7.93)
PET3 11 11 1.1 1.7 1.60 (0.65-3.92)
Mean
PET2 10 10 1.1 1.7 1.21 (0.49-2.99)
PET3 11 11 1.1 1.9 1.58 (0.64-3.88)
Hetero
PET2 10 10 1.0 1.9 2.68 (1.00-7.21)
PET3 11 11 1.1 2.1 4.21 (1.28-13.8)
Favors ≥ median
Favors < median

Abbreviations: PET, positron emission tomography; PET2, week 13; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression; PSA, prostate-specific antigen; SUV, standardized uptake value.

Figure 7. Time to (A) PSA Progression and (B) Treatment Discontinuation by Change in SUV_{hetero} From Baseline for All Bone Lesions at PET3

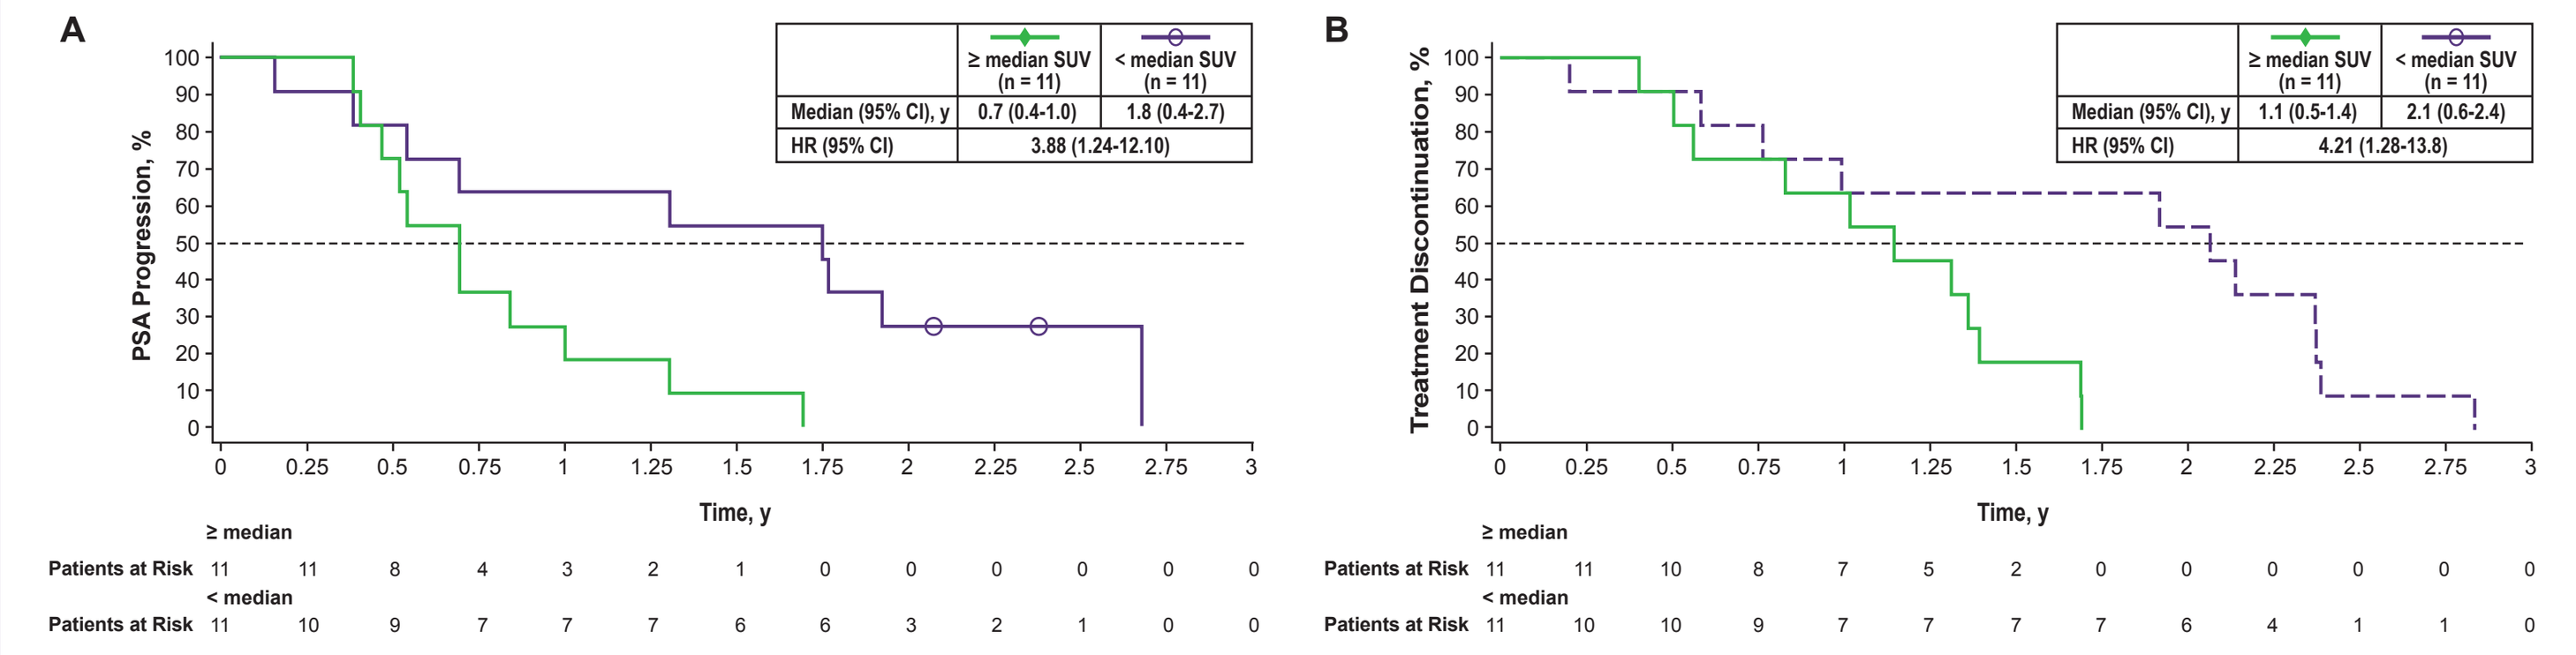
A
≥ median SUV (n = 11)
< median SUV (n = 11)
Median (95% CI), y 0.7 (0.4-1.0) 1.8 (0.4-2.7)
HR (95% CI) 3.88 (1.24-12.10)
PSA Progression, %
Time, y
≥ median Patients at Risk 11 11 8 4 3 2 1 0 0 0 0 0 0
< median Patients at Risk 11 10 9 7 7 7 6 6 3 2 1 0 0
B
≥ median SUV (n = 11)
< median SUV (n = 11)
Median (95% CI), y 1.1 (0.5-1.4) 2.1 (0.6-2.4)
HR (95% CI) 4.21 (1.28-13.8)
Treatment Discontinuation, %
Time, y
≥ median Patients at Risk 11 11 10 8 7 5 2 0 0 0 0 0 0
< median Patients at Risk 11 10 10 9 7 7 7 7 6 4 1 1 0

Hazard ratio is relative to < median group, with values > 1 indicating more risk in the ≥ median group.
Abbreviations: HR, hazard ratio; PET, positron emission tomography; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression; PSA, prostate-specific antigen; SUV, standardized uptake value; SUV_{hetero}, heterogeneity of response across all bone lesions.

- In an individual patient, prolonged duration of treatment is reflected by a decrease in SUV_{hetero} (**Figure 8**).

Figure 8. Examples of Histograms Showing Change in SUV_{hetero} at Baseline (PET1) and PET3

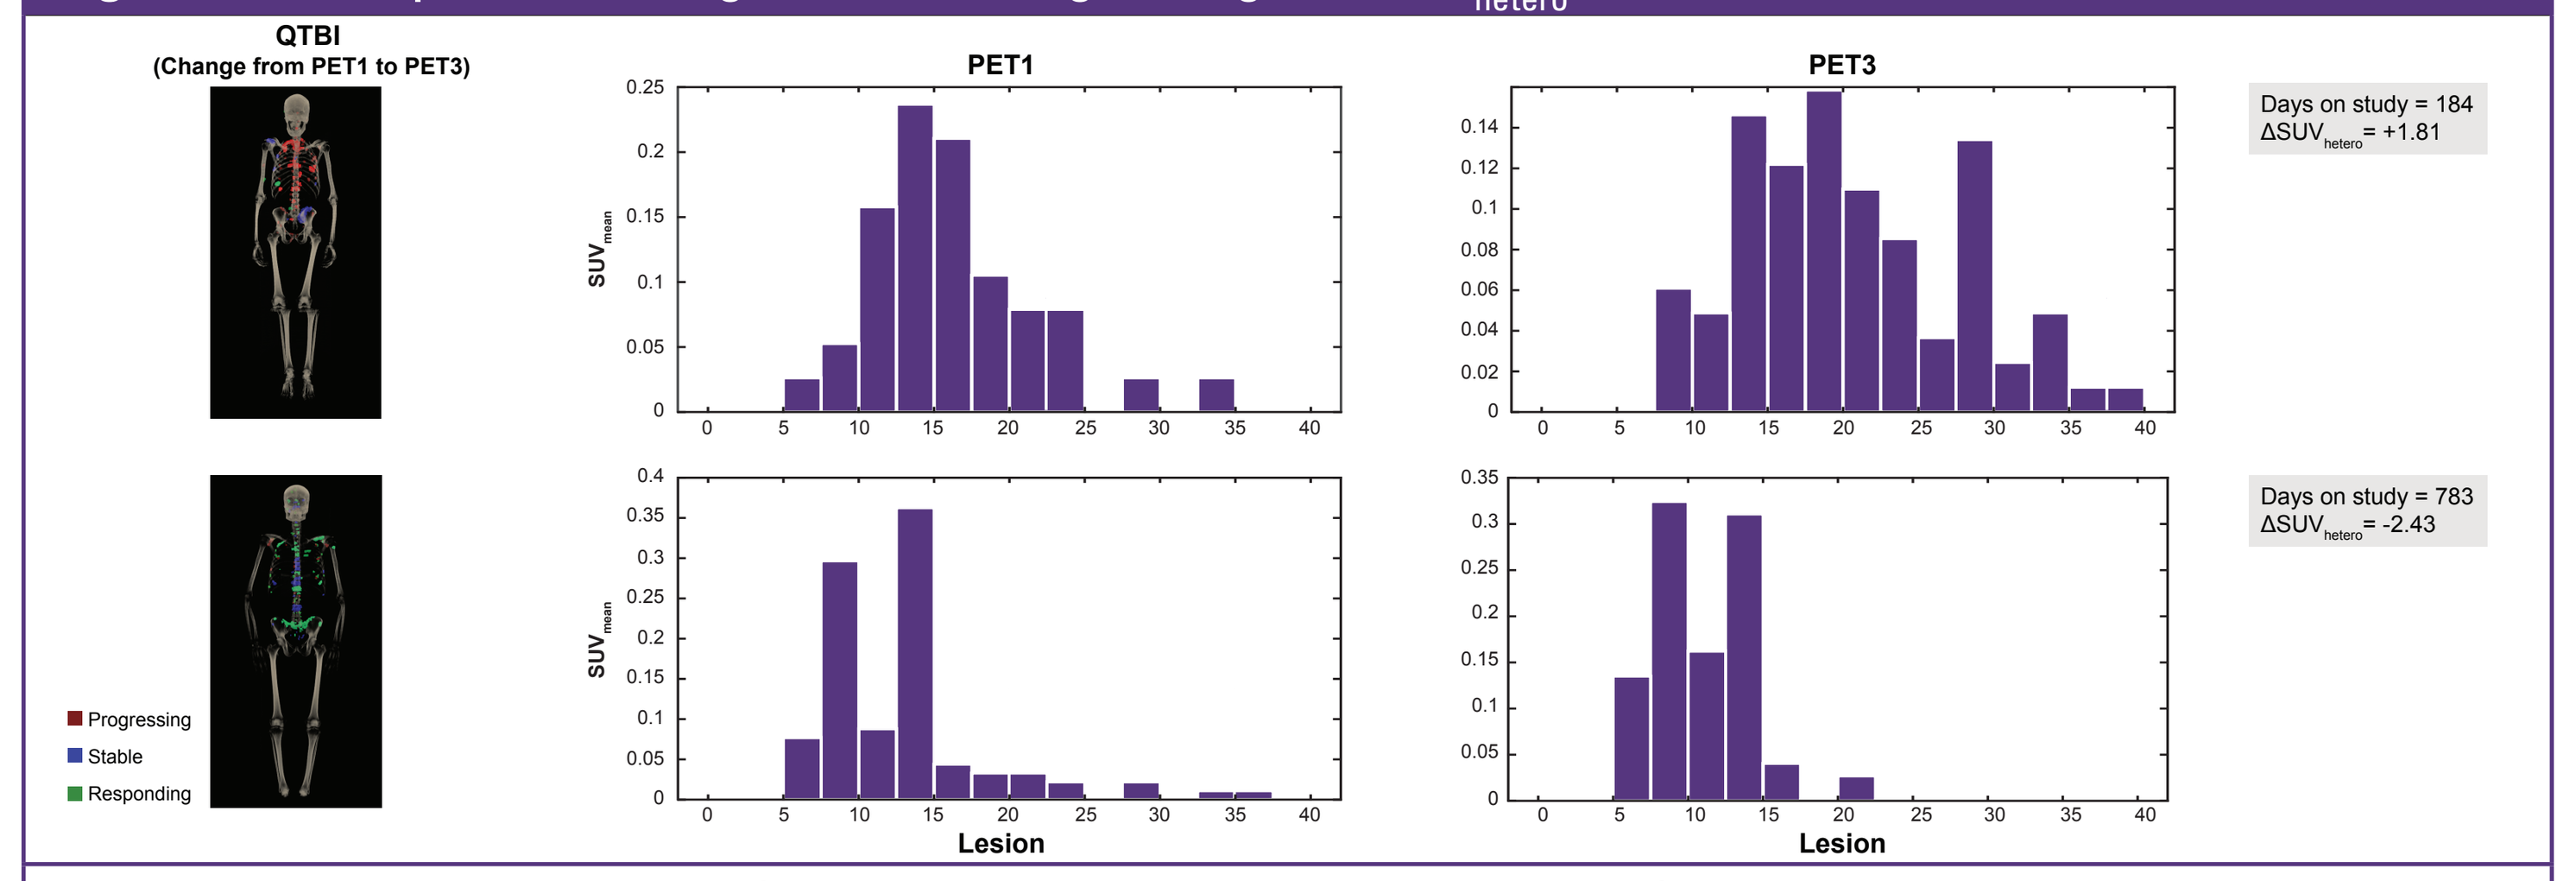
QTBI (Change from PET1 to PET3)
PET1
PET3
SUV_{mean}
Lesion
Days on study = 184
ΔSUV_{hetero} = +1.81
Days on study = 783
ΔSUV_{hetero} = -2.43
Progressing
Stable
Responding

Abbreviations: PET, positron emission tomography; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression; QTBI, quantitative total bone imaging; SUV, standardized uptake value; SUV_{hetero}, heterogeneity of response across all bone lesions; SUV_{mean}, the average activity of all lesions.

Evaluation of CTCs

- Expression of androgen receptor splice variants and neuroendocrine genes in CTCs increased after treatment with enzalutamide (**Figure 9**).
 - Prior to treatment, 35% of patients had CTCs expressing androgen-receptor splice variant 7 (AR-V7) or synaptophysin (a neuroendocrine marker); after treatment, this percentage of patients increased to 63%.

Figure 9. CTC Analysis (A) Prior to Treatment and (B) Heatmap of Expression of Individual Genes

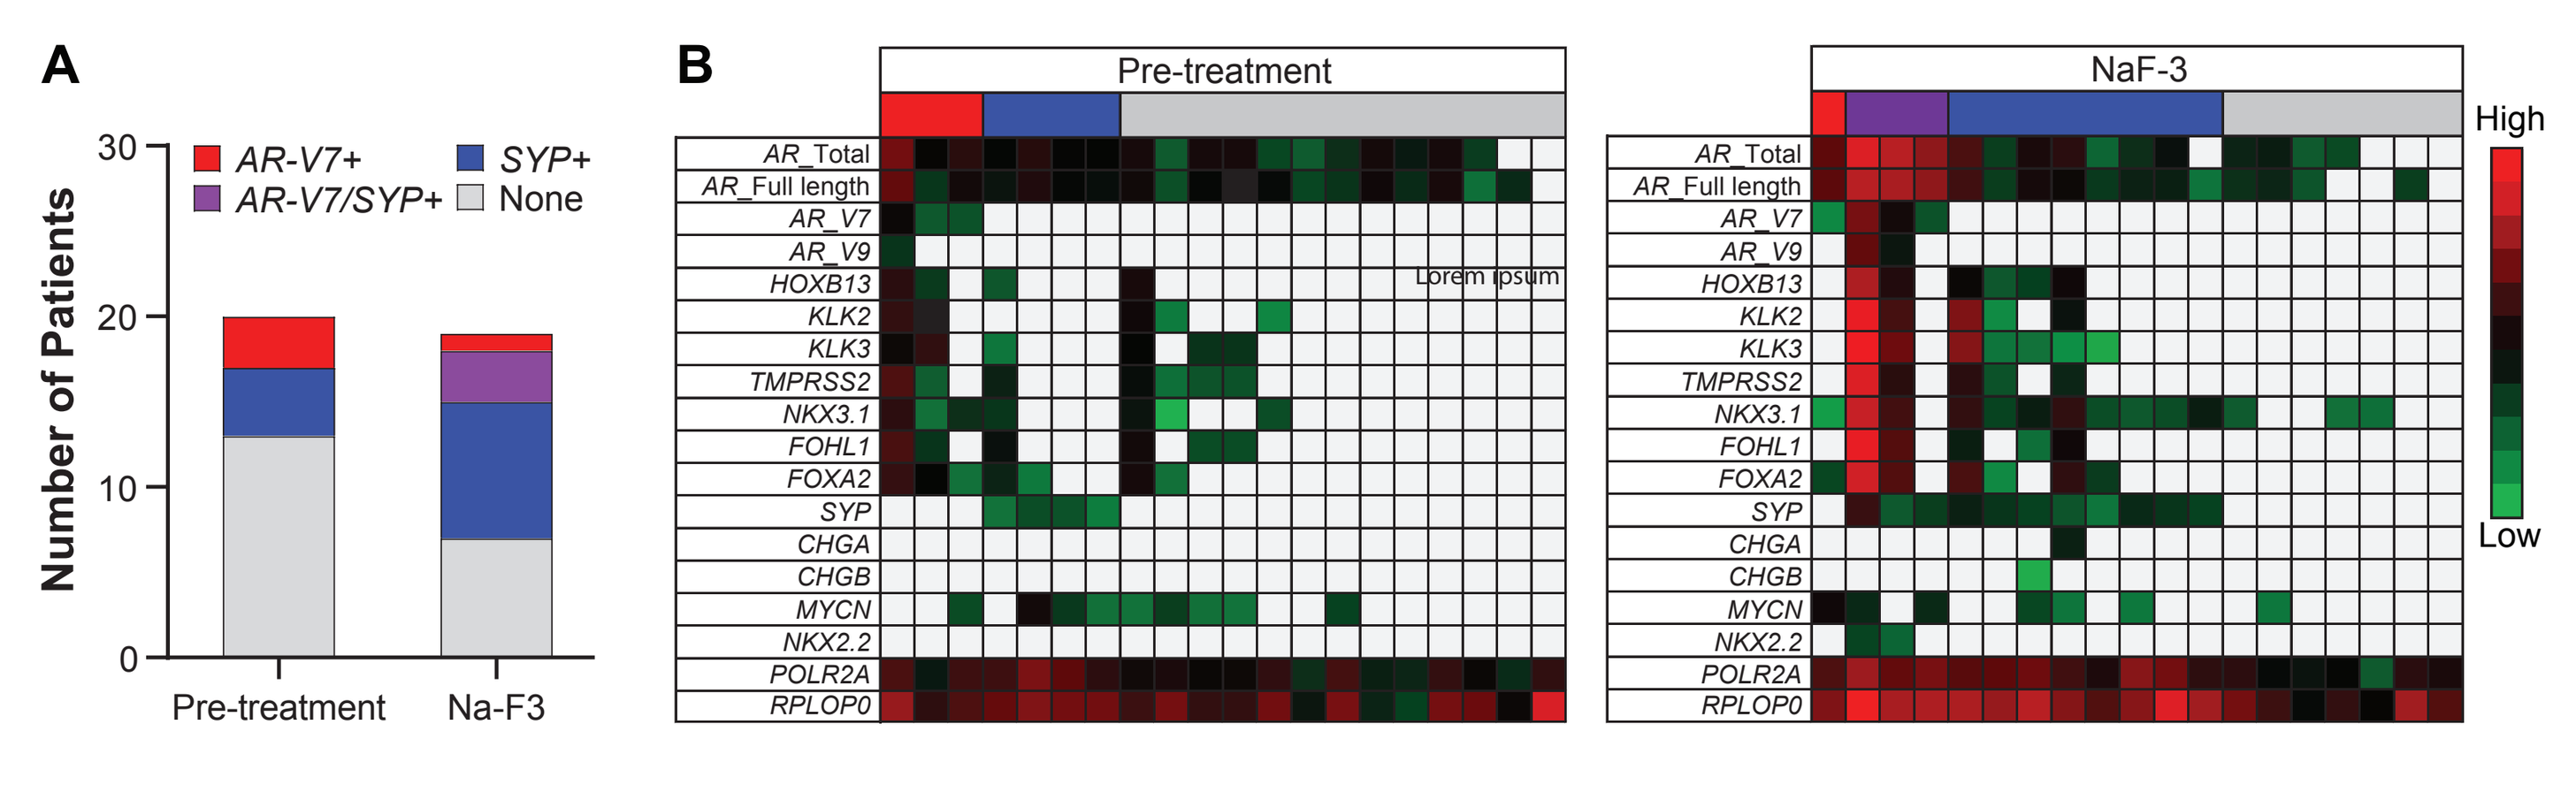
A
AR-V7+
AR-V7/SYP+
SYP+
None
Number of Patients
Pre-treatment
Na-F3
B
Pre-treatment
NaF-3
AR_Total
AR_Full length
AR_V7
AR_V9
HOXB13
KLK2
KLK3
TMPRSS2
NKX3.1
FOHL1
FOXA2
SYP
CHGA
CHGB
MYCN
NKX2.2
POLR2A
RPLOP0
High
Low

Abbreviations: AR, androgen receptor; AR-V, AR splice variant; CTC, circulating tumor cell; SYP, synaptophysin; PET, positron emission tomography; PET3, time of PSA, standard radiographic, or clinical progression or at 2 years without progression.

Safety

- Enzalutamide was generally well tolerated. The most common adverse events were fatigue (82.6%), arthralgia (26.1%), back pain (26.1%), diarrhea (26.1%), and falls (26.1%).

CONCLUSIONS

- We found that duration of enzalutamide treatment was not associated with baseline tumor burden, as measured by SUV_{total}, but that a decrease at week 13 in tumor heterogeneity, as measured by change in SUV_{hetero} from baseline, was associated with longer duration of treatment and longer time to PSA progression.
- Although baseline heterogeneity may be prognostic, it may not predict response or duration of therapy.
- Treatment response heterogeneity may predict benefit duration and should be explored as a possible surrogate in patients with prostate cancer.
- AR-V7/9 or neuroendocrine expression in CTCs did not predict lack of benefit from enzalutamide; however, at the time of PET3, the observed increase in AR-V7/9 and neuroendocrine expression are consistent with tumor evolution to more aggressive phenotypes.
- Treatment with enzalutamide beyond progression may be an option in some patients.
 - In men with oligoresistant disease, early ablation of nonresponding lesions may be of clinical benefit to maximize treatment response and delay progression.
 - In men with oligoprogressive disease at the time of PSA progression, identification and ablation of progressive lesions may extend clinical benefit with enzalutamide.

REFERENCES

1. Dagogo-Jack I, Shaw AT. *Nat Rev Clin Oncol.* 2018;15(2):81-94.
2. Kyriakopoulos C, et al. *J Clin Oncol.* 2019;37(15_suppl):5036.
3. Scher HI, et al. *J Clin Oncol.* 2008;26(7):1148-1159.

ACKNOWLEDGMENTS

This study was sponsored by Pfizer Inc. (New York, NY) and Astellas Pharma, Inc. (Northbrook, IL), the co-developers of enzalutamide. The data presented in Figures 8 and 9 were generated, validated, and are stored by AIQ Solutions (Madison, WI). Medical writing and editorial assistance funded by both sponsor companies was provided by Stephanie Vadasz, PhD, and Mary Kacillas from Ashfield Healthcare Communications.

DISCLOSURES

GL is an employee of, has a leadership role at, and owns stock or has other ownership interests in AIQ Solutions; has served as a consultant for Novartis, Exelixis, Janssen, and TRACON Pharma; holds an institutional patent; has received institutional funding from Johnson & Johnson and Novartis; and has received research funding from Madison Vaccines, Inc., Pfizer, and TRACON Pharma. Disclosures for the remaining authors can be obtained through the Quick Response (QR) link containing the poster.

Please scan this QR code with your smartphone app to view an electronic version of this poster. If you do not have access to a smartphone, please access the poster via the following link: https://congress-download.pfizer.com/esmo_2019_european_society_of_medical_oncology_511_enzalutamide_liu_g_2226.html
Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors. Please contact gxl@medicine.wisc.edu for more information regarding this poster.

PLAIN LANGUAGE SUMMARY
Please scan this quick response (QR) code with your smartphone app to view a plain language summary.